AML-229

Efficacy and safety of pivekimab sunirine in patients with blastic plasmacytoid dendritic cell neoplasm in the CADENZA study

Naveen Pemmaraju,[1] Giovanni Marconi,[2] Pau Montesinos,[3] Andrew A. Lane,[4] Luca Mazzarella,[5] David A. Sallman,[6] Matthew L. Ulrickson,[7] Gary J. Schiller,[8] Harry Paul Erba,[9] Eunice S. Wang,[10] Roland B.Walter,[11] Eric Deconinck,[12] Ahmed M. Aribi,[13] Ollivier Legrand,[14] Delphine Lebon,[15] Giovanni Martinelli,[2] Daniel J. DeAngelo,[4] Yining Du,[16] Jalaja Potluri,[16] Naval Guastad Daver[1]

[1]MD Anderson Cancer Center, Houston, TX, USA; [2]Istituto Scientifico Romagnolo per lo Studio e la Cura dei Tumori "Dino Amadori" (IRST) IRCCS, Gatteo, Italy; [3]Hospital Universitari i Politècnic La Fe, Valencia, Spain; [4]Dana-Farber Cancer Institute, Boston, MA, USA; [5]IEO, European Institute of Oncology IRCCS, Milano, Italy; [6]Moffitt Cancer Center and Research Institute, Tampa, FL, USA; [7]Banner MD Anderson Cancer Center, Gilbert, AZ, USA; [8]David Geffen School of Medicine at UCLA, Los Angeles, CA, USA; [9]Duke University, Durham, NC, USA; [10]Roswell Park Comprehensive Cancer Center, Buffalo, NY, USA; [11]Fred Hutchinson Cancer Center, Seattle, WA, USA; [12]CHU Besançon-Hématologie, Besançon, France; [13]City of Hope, Duarte, CA, USA; [14]Hospital St. Antoine, Paris, France; [15]CHU Picardie, Amiens, France; [16]AbbVie, Inc., North Chicago, IL, USA

of Genmab at the time the research was conducted

OBJECTIVE

To evaluate the efficacy and safety of PVEK in patients with BPDCN

CONCLUSIONS

PVEK was associated with high and durable responses in 1L (de novo and PCHM) patients with BPDCN

- CR + CRc = 70% (95% CI: 51.3–84.4)

PVEK provides early and durable CR + CRc responses for R/R patients (median 1.4 and 9.2 months, respectively)

PCHM did not impact OS in 1L patients

- 16.6 (7.2, NR) versus 16.6 (11.4, NR) months in patients with de novo and PCHM BPDCN, respectively

~One half of 1L patients treated with PVEK subsequently received SCT

PVEK treatment had a manageable safety profile;

- Infusion-related reactions and peripheral edema were resolved with supportive care
- No capillary-leak syndrome observed

BACKGROUND

- BPDCN is a rare, aggressive hematological malignancy primarily involving skin, bone marrow, lymph nodes, and CNS[1]
- Current treatment options are limited to chemotherapy regimens adapted from AML, ALL, lymphoma and myeloma
 - o Tagraxofusp, an IL-3Rα CD123-directed cytotoxin agent (only approved treatment for BPDCN)[1,2]
- Median age of BPDCN diagnosis is ~65 years; many patients are not eligible for intensive chemotherapy and precluded from SCT[3]
- CD123 (IL-3Rα) is overexpressed on all BPDCN blasts, making it an ideal target for immunochemotherapy[4]

[a]Category 2a per National Comprehensive Cancer Network guidelines. Referenced with permission from the NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines® for B-Cell Lymphomas V1.2024). ©National Comprehensive Cancer Network, Inc. 2024. All rights reserved. Accessed March 5, 2025. To view the most current and complete version of the guideline, go online to NCCN.org.

PVEK structure and mechanism of action

PVEK structure

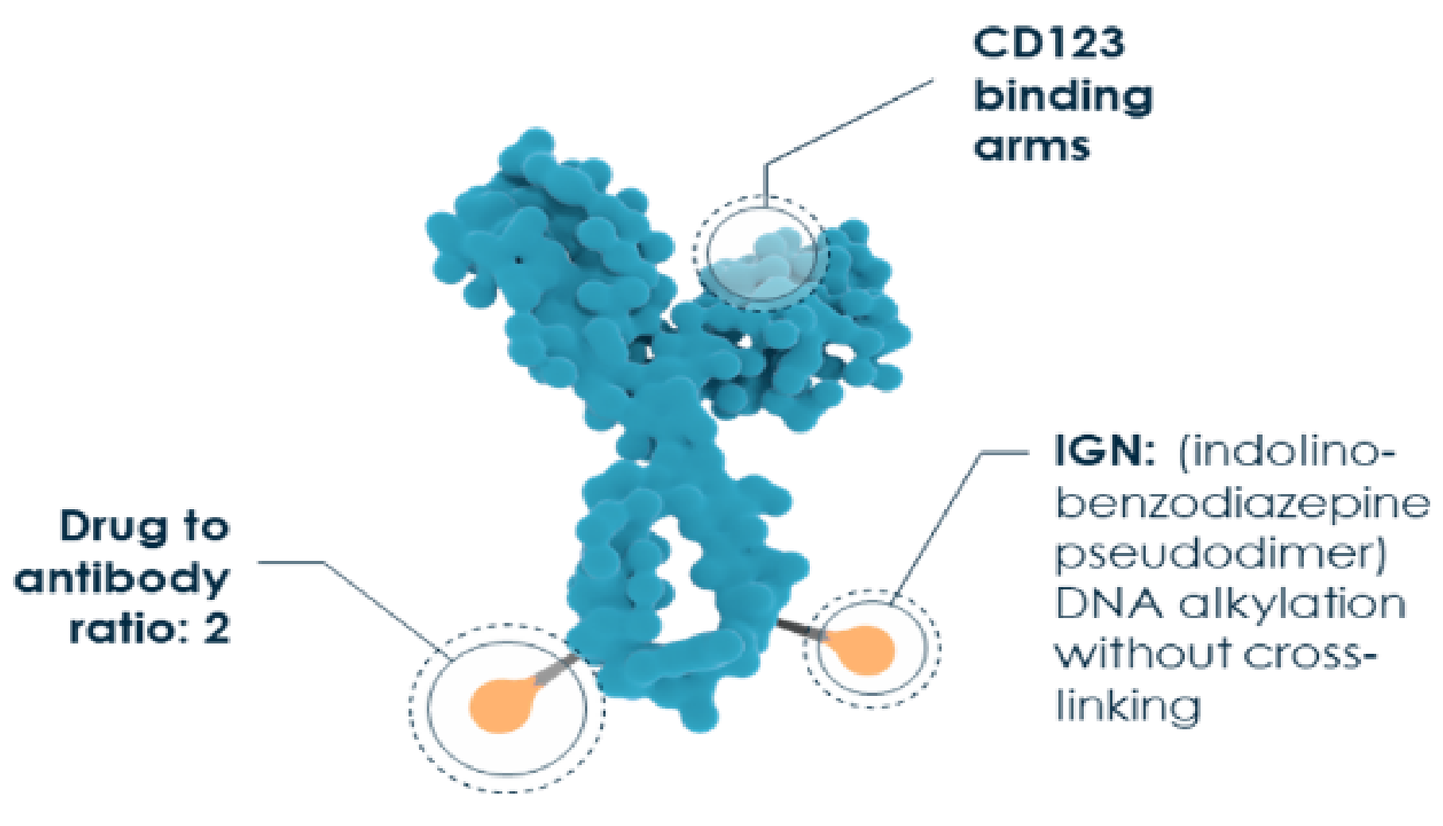
CD123 binding arms
Drug to antibody ratio: 2
IGN: (indolino-benzodiazepine pseudodimer) DNA alkylation without cross-linking

PVEK mechanism of action

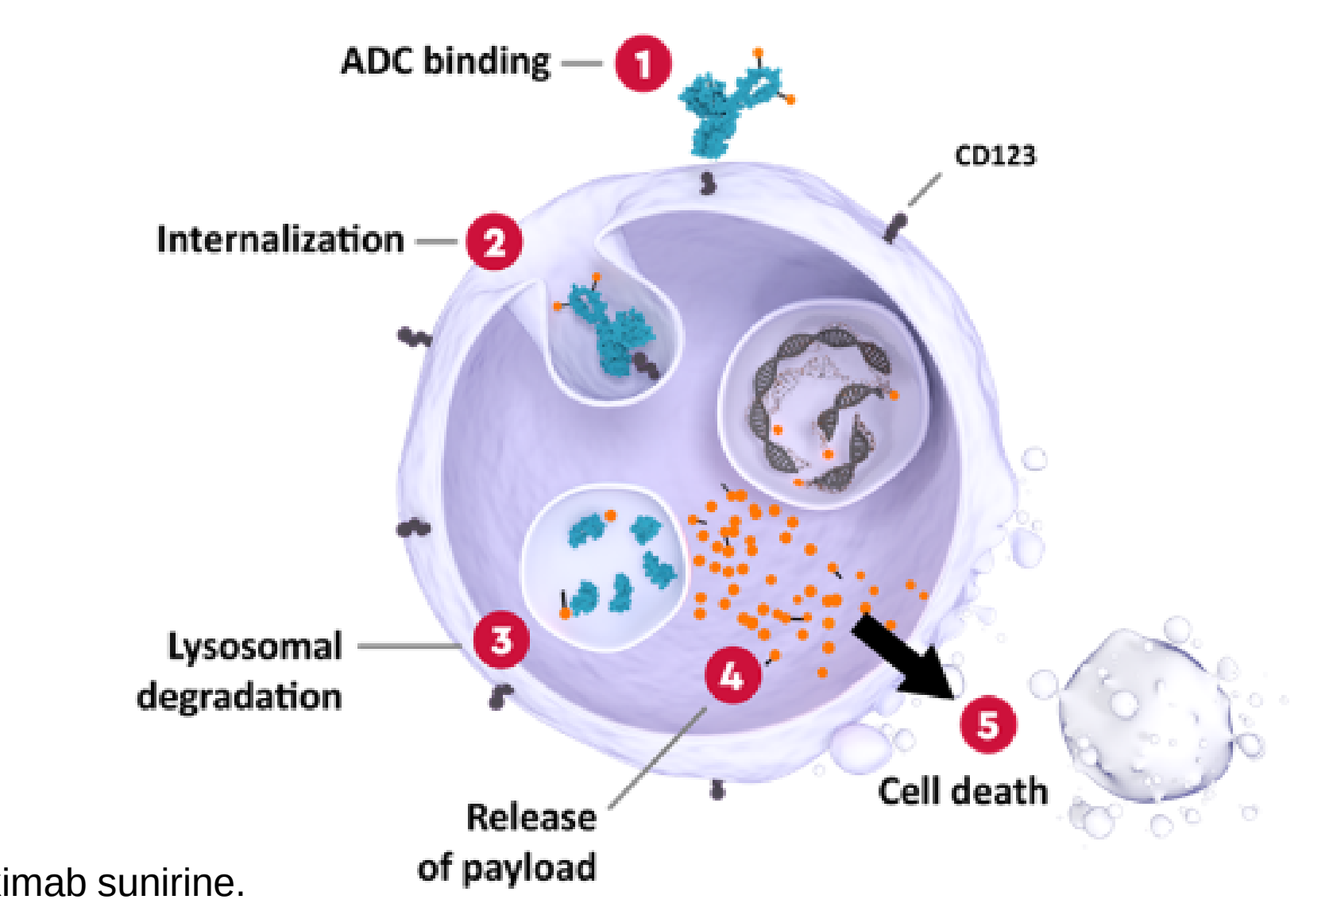
ADC binding 1
CD123
Internalization 2
Lysosomal degradation 3
4
Release of payload
5
Cell death

ADC, antibody-drug conjugate; IGN, indolinobenzodiazepine pseudodimer; PVEK, pivekimab sunirine.

STUDY DESIGN

CADENZA: Nonrandomized, open-label, multicenter, Phase 1/2 study in patients with CD123-positive BPDCN

Two patient groups

- **No prior exposure to systemic therapy (1L)**
 - De novo BPDCN (**Primary analysis population[a]**)
 - BPDCN with prior or concomitant hematologic malignancy (PCHM)
- **R/R BPDCN (1–3 prior systemic therapies)**

All patients received PVEK intravenously (<30-min infusion) on **Day 1 of a 21-day cycle** at the recommended phase 2 dose of **0.045 mg/kg**

Exclusion criteria

✓ ECOG PS score of ≤1
✓ Adequate organ function and liver enzyme range
✓ Aged ≥18 years

Eligibility criteria

X History of hepatic veno-occlusive disease, grade 4 capillary-leak syndrome, or noncardiac grade 4 edema
X 1L patients with central nervous system disease[b]

[a]The primary analysis population included 1L patients with no PCHM, who met eligibility criteria and enrolled following the protocol amendment to define the de novo population, and received at least 1 dose of PVEK; [b]Patients with R/R disease with central nervous system disease could be included if treated and had no evidence of central nervous system disease before first treatment dose.1L, frontline; BPDCN, blastic plasmacytoid dendritic cell neoplasm; ECOG, Eastern Cooperative Oncology Group; PCHM, prior or concomitant hematologic malignancy; PS, performance status; PVEK, pivekimab sunirine; R/R, relapsed or refractory.

Study endpoints

Primary: Rate of composite CR, defined as CR[a] + clinical CR (CR with minimal skin abnormality [CRc]) in 1L de novo patients

Key secondary:

- Duration of CR + CRc in **1L de novo patients**

Additional secondary:

- CR+CRc rate
- Time to CR+CRc
- ORR[b]
- OS
- Percentage of patients bridged to posttreatment SCT
- Frequency and severity of adverse events

In **overall 1L group** (de novo/PCHM cohorts) and **R/R group**

[a]Complete response required: Normalization of blast percentage and neutrophil and platelet counts, absence of leukemic blasts, complete clearance of skin lesions and no new lesions in patients without lesions at baseline, regression of nodal masses to normal size on computed tomography, and no splenic or hepatic nodules; [b]ORR was defined as CR + CRc + CRh + CRi + PR. 1L, frontline; CR, complete response; CRc, CR with minimal skin abnormality; CRh, complete remission with partial hematologic recovery; CRi, complete remission with incomplete recovery; ORR, overall response rate; OS, overall survival; PCHM, prior or concomitant hematologic malignancy; PR, partial response; R/R, relapsed or refractory; SCT, stem cell transplant.

RESULTS

Enrolled 84 patients with BPDCN

Patient disposition

1L group (N=33[a])
- De novo BPDCN (n=22)
 - o Primary analysis population[b] (n=20)
- PCHM BPDCN (n=11)

Primary reason for discontinuing treatment
- 12 progressive disease
- **12 bridging to transplant**
- 3 AE
- 2 death
- 1 clinical progression
- 1 withdrew consent
- 1 investigator decision[c]

R/R group (N=51[a])

Primary reason for discontinuing treatment
- 30 progressive disease
- **6 bridging to transplant**
- 3 AEs
- 5 clinical progression
- 2 withdrew consent
- 4 investigator decision[c]

Data cutoff: October 2, 2024

[a]One patient from the 1L group and one patient from the R/R group were still on treatment at the time of data cutoff; [b]The primary analysis population included frontline patients with no PCHM, who met eligibility criteria and enrolled following the protocol amendment to define the de novo population and received at least 1 dose of PVEK; [c]Not related to an AE. 1L, frontline; AE, adverse event; BPDCN, blastic plasmacytoid dendritic cell neoplasm; PCHM, prior or concomitant hematologic malignancy; PVEK, pivekimab sunirine; R/R, relapsed or refractory.

Baseline demographics and clinical characteristics

Characteristic	1L de novo BPDCN (n=20)[a]	1L (de novo and PCHM) BPDCN (n=33)[b]	R/R BPDCN (n=51)
Age, years, median (range)	74 (48–84)	73 (48–84)	69 (19–85)
Aged ≥65 years	17 (85.0)	30 (90.9)	30 (58.8)
Male	17 (85.0)	27 (81.8)	42 (82.4)
Race			
White	15 (75.0)	27 (81.8)	42 (82.4)
Hispanic/Latino	3 (15.0)	4 (12.1)	8 (15.7)
Black	1 (5.0)	1 (3.0)	2 (3.9)
Asian	0 (0)	0 (0)	1 (2.0)
Not reported	4 (20.0)	5 (15.2)	6 (11.8)
ECOG PS			
0	7 (35.0)	12 (36.4)	18 (35.3)
1	13 (65.0)	21 (63.6)	30 (58.8)
2	0 (0)	0 (0)	2 (3.9)
3	0 (0)	0 (0)	1 (2.0)
Disease involvement			
Skin	18 (90.0)	31 (93.9)	34 (66.7)
Bone marrow	9 (45.0)	16 (48.5)	24 (47.1)
Peripheral blood	1 (5.0)	3 (9.1)	10 (19.6)
Lymph nodes	6 (30.0)	12 (36.4)	18 (35.3)
Viscera	0 (0)	0 (0)	3 (5.9)
Disease history/type			
De novo	20 (100)	22 (66.7)	42 (82.4)
Secondary (PCHM)	0 (0)	11 (33.3)	9 (17.6)

Characteristic	R/R BPDCN (n=51)
Status at baseline	
First relapse	15 (29.4)
Primary refractory	25 (49.0)
Relapsed refractory	11 (21.6)
Prior therapies	
Nonintensive chemotherapy	32 (62.7)
Tagraxofusp	29 (56.9)
Intensive chemotherapy	25 (49.0)
Prior HSCT	16 (31.4)
Radiotherapy	9 (17.6)
Prior lines of therapy[c]	
1	29 (56.9)
2	12 (23.5)
≥3	9 (17.6)

Data are presented as n (%) unless otherwise indicated. [a]Primary analysis population including patients who met eligibility criteria, prospectively enrolled, and received at least 1 dose of PVEK. These patients were a subset of the frontline BPDCN group; [b]Includes 22 patients with de novo BPDCN (20 patients in the primary analysis plus 2 patients who enrolled early) and 11 patients with PCHM malignancy; [c]Data on prior therapy was missing for one patient.1L, frontline; BPDCN, blastic plasmacytoid dendritic cell neoplasm; ECOG, Eastern Cooperative Oncology Group; HSCT, hematopoietic stem-cell transplant; PCHM, prior or concomitant hematologic malignancy PS, performance status; PVEK, pivekimab sunirine; R/R, relapsed or refractory.

Response rates in patients with BPDCN

Composite CR and ORR

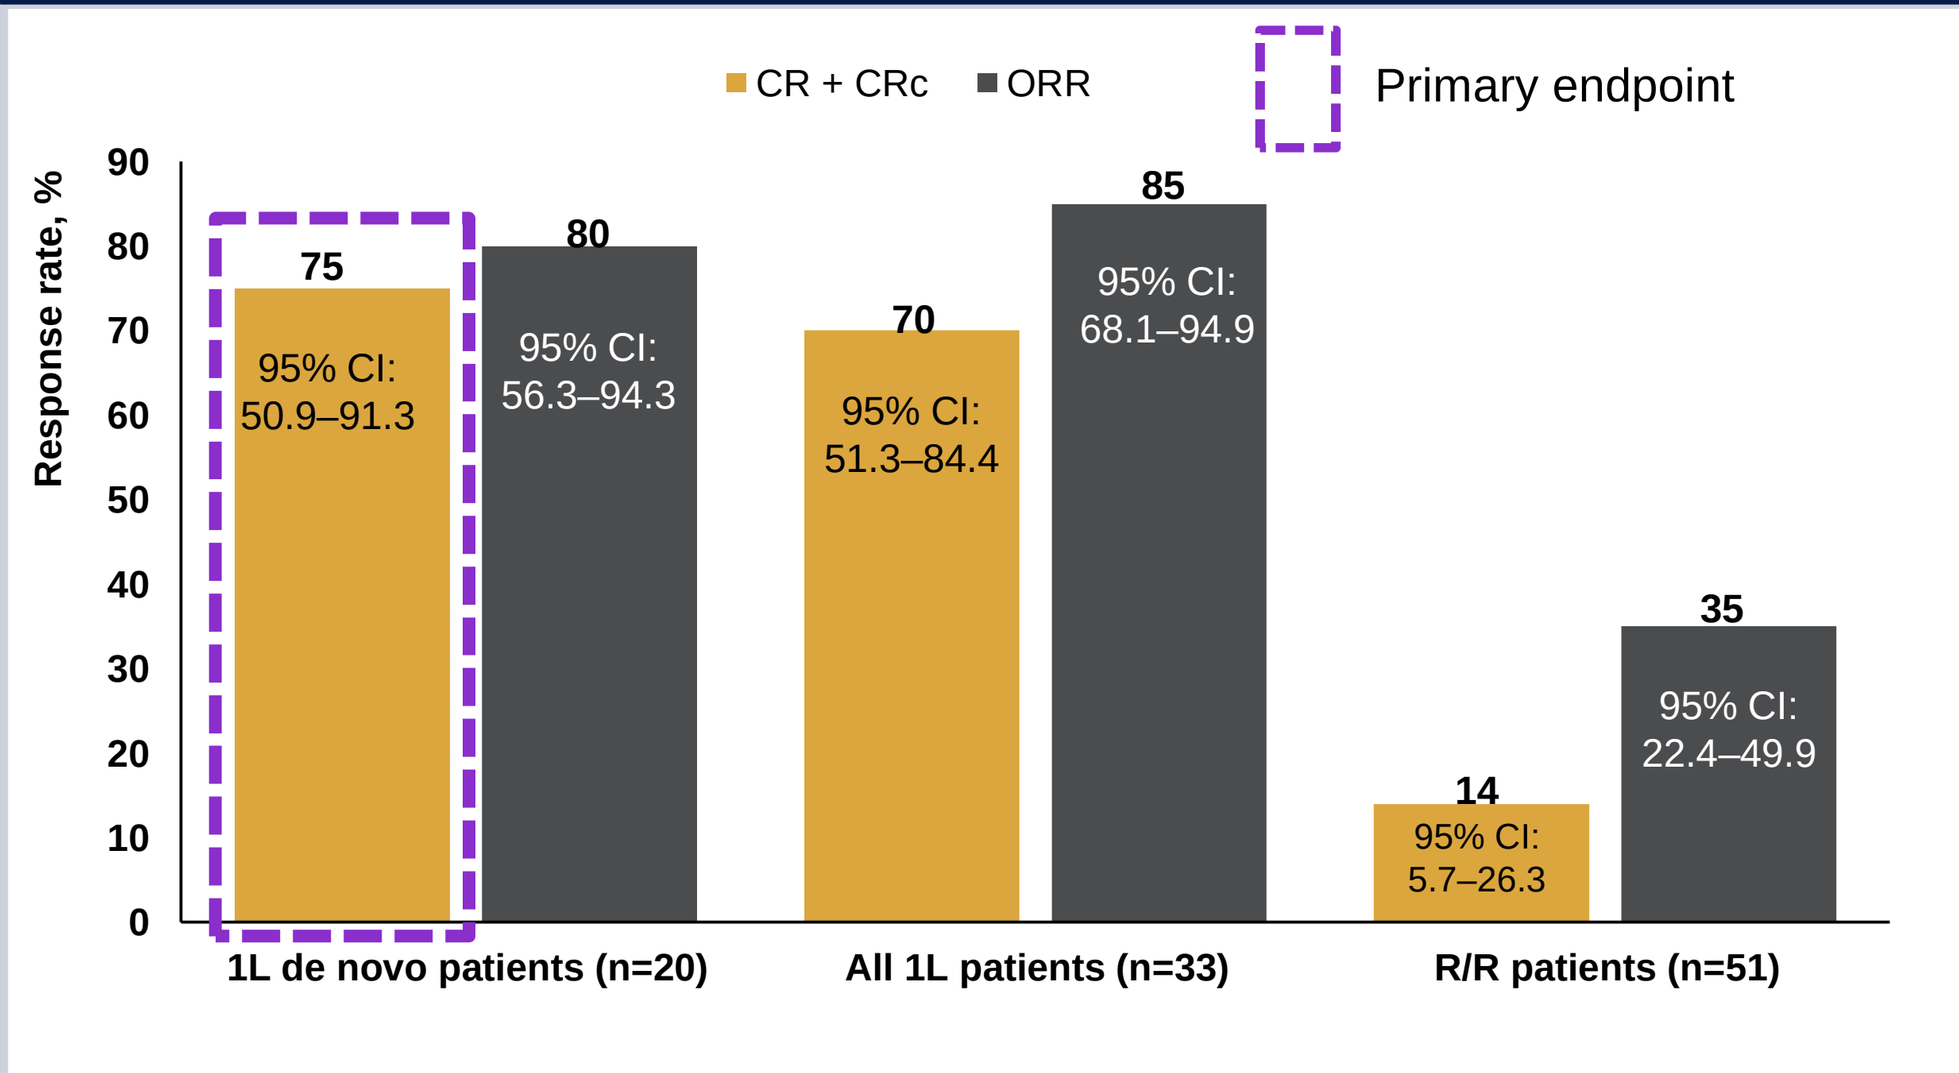
CR + CRc
ORR
Primary endpoint
Response rate, %
75
95% CI: 50.9–91.3
80
95% CI: 56.3–94.3
70
95% CI: 51.3–84.4
85
95% CI: 68.1–94.9
14
95% CI: 5.7–26.3
35
95% CI: 22.4–49.9
1L de novo patients (n=20)
All 1L patients (n=33)
R/R patients (n=51)

	1L de novo BPDCN (n=20)	1L (de novo and PCHM) BPDCN (n=33)	R/R BPDCN (n=51)
Median duration of CR + CRc, months (95% CI)	10.6 (3.8–NR)	9.8 (4.6–NR)	9.2 (2.4–NR)
Median time to CR + CRc, months (interquartile range)	1.4 (1–3)	1.5 (1–3)	1.4 (1–2)

1L, frontline; BPDCN, blastic plasmacytoid dendritic cell neoplasm; CI, confidence interval; CR, complete response; CRc, CR with minimal skin abnormality; NR, not reached; ORR, overall response rate; OS, overall survival; PCHM, prior or concomitant hematologic malignancy; PVEK, pivekimab sunirine; R/R, relapsed or refractory; SCT, stem cell transplant.

Response rates were similar between R/R patients **with** (n=29) and **without** (n=22) prior tagraxofusp exposure

- **CR + CRc**: 14% (95% CI:4–32) and 14% (95% CI:3–35)
- **ORR:** 35% (95% CI:18–54) and 36% (95% CI:17–59)

OS was similar between patients with de novo and PCHM BPDCN

	1L de novo BPDCN (n=20)	1L (de novo and PCHM) BPDCN (n=33)	R/R BPDCN (n=51)
Median OS, months (95% CI)	16.6 (7.2, NR)	16.6 (11.4, NR)	5.8 (3.9–8.4)

OS in all 1L patients (de novo and PCHM) BPDCN

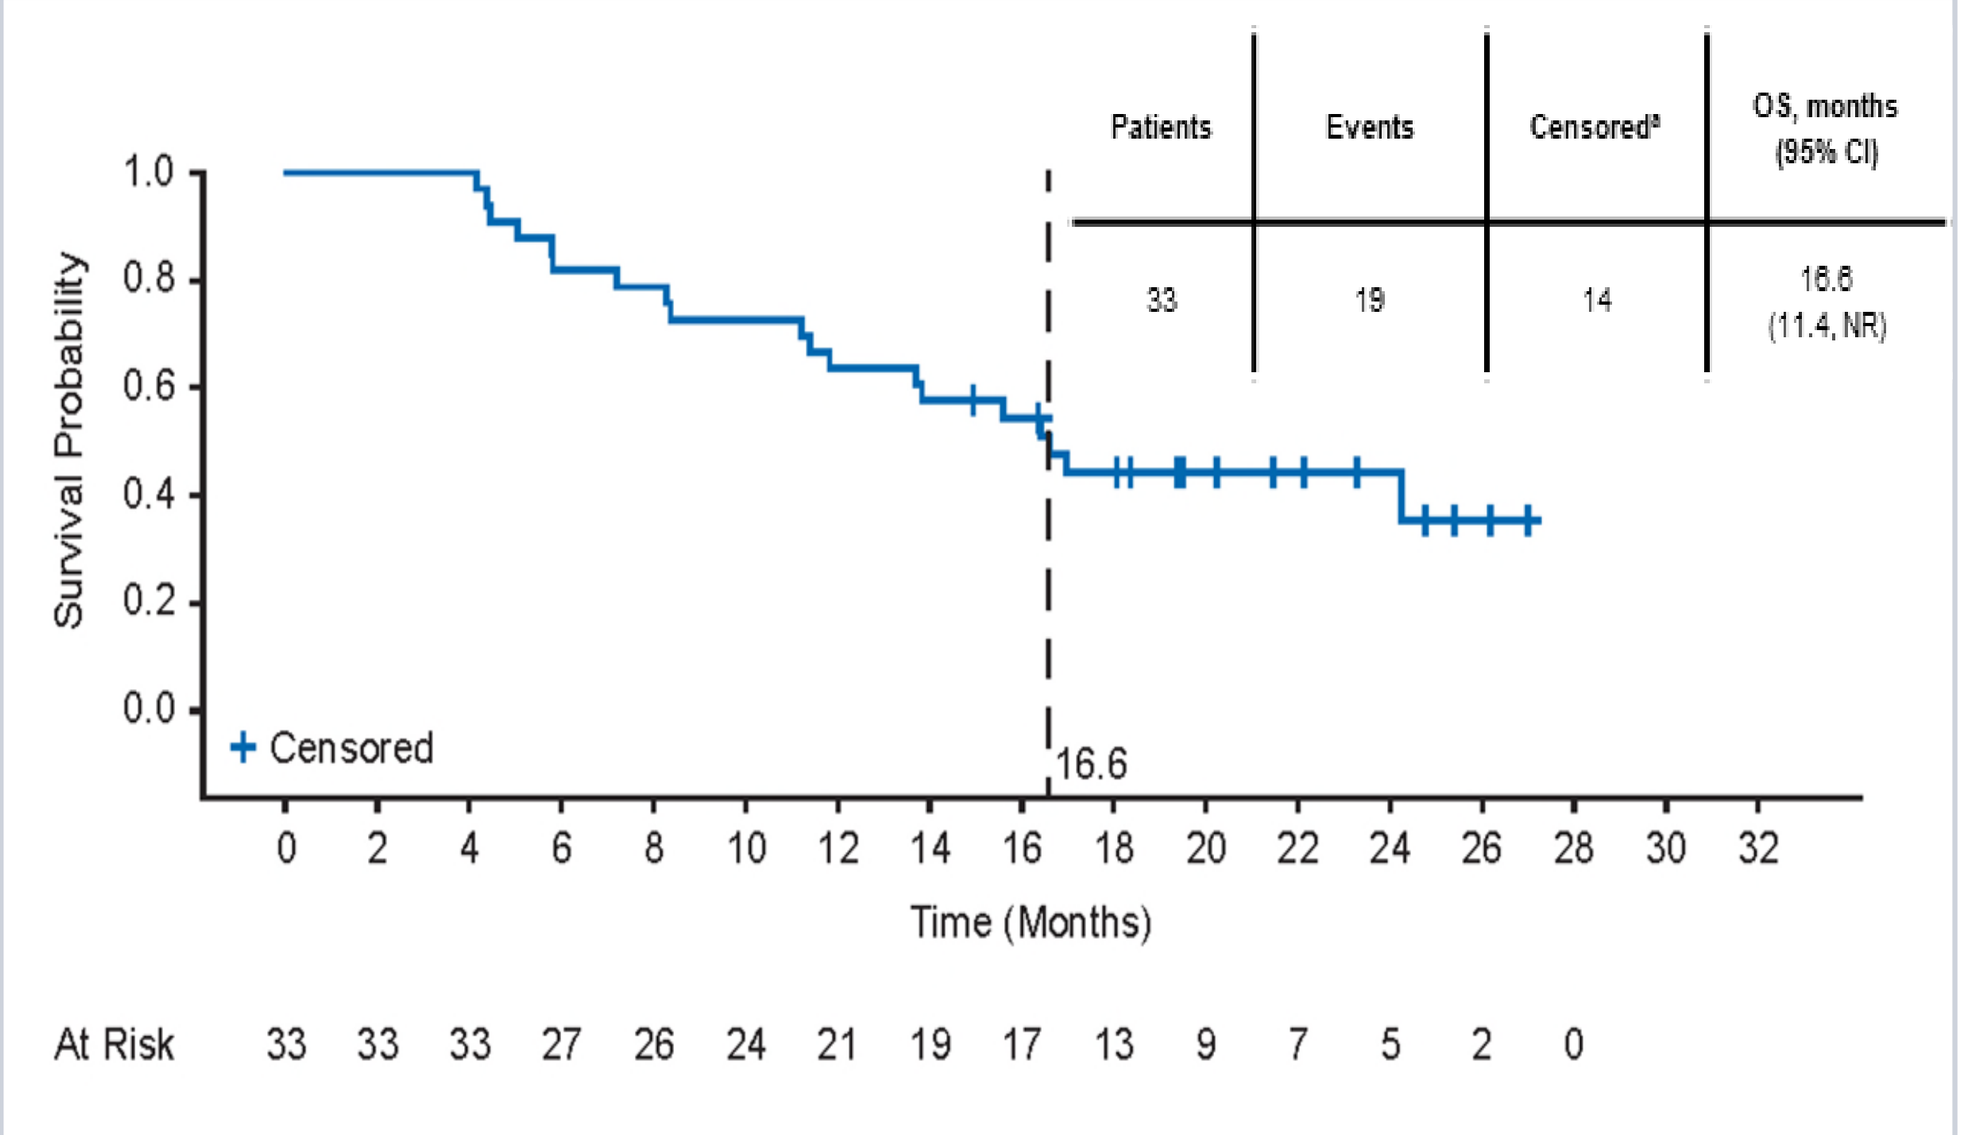
Patients
Events
Censored[a]
OS, months (95% CI)
33
19
14
16.6 (11.4, NR)
Survival Probability
Censored
16.6
Time (Months)
At Risk 33 33 33 27 26 24 21 19 17 13 9 7 5 2 0

[a]In the OS analysis, patients who were not confirmed to have died at time of analysis were censored on the last known alive date. 1L, frontline; BPDCN, blastic plasmacytoid dendritic cell neoplasm; CI, confidence interval; NR, not reached; OS, overall survival; PCHM, prior or concomitant hematologic malignancy; PVEK, pivekimab sunirine; R/R, relapsed or refractory; SCT, stem cell transplant.

PVEK treatment enables SCT with OS benefit in all patients

52% of all 1L responders (CR +CRc) received SCT

OS by SCT status (all 1L; de novo and PCHM)

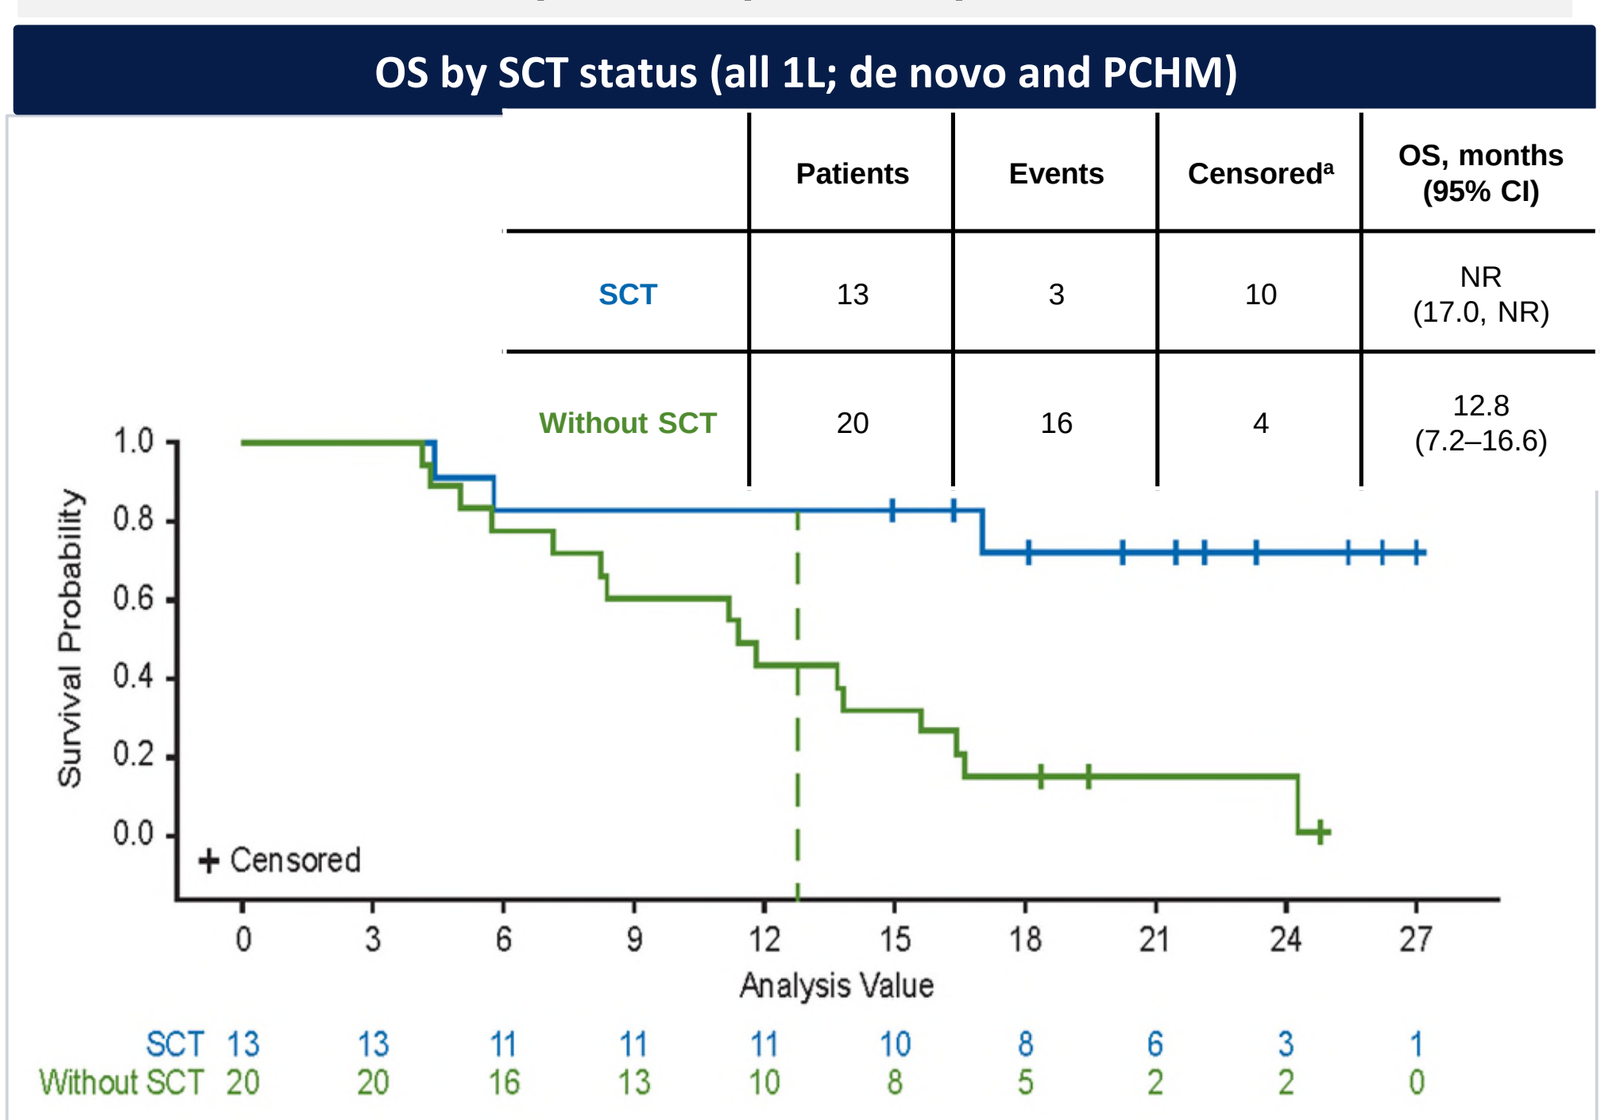
Patients
Events
Censored[a]
OS, months (95% CI)
SCT 13 3 10 NR (17.0, NR)
Without SCT 20 16 4 12.8 (7.2–16.6)
Survival Probability
Censored
Analysis Value
SCT 13 13 11 11 11 10 8 6 3 1
Without SCT 20 20 16 13 10 8 5 2 2 0

29% of R/R responders (CR + CRc) received SCT

OS by SCT status (R/R)

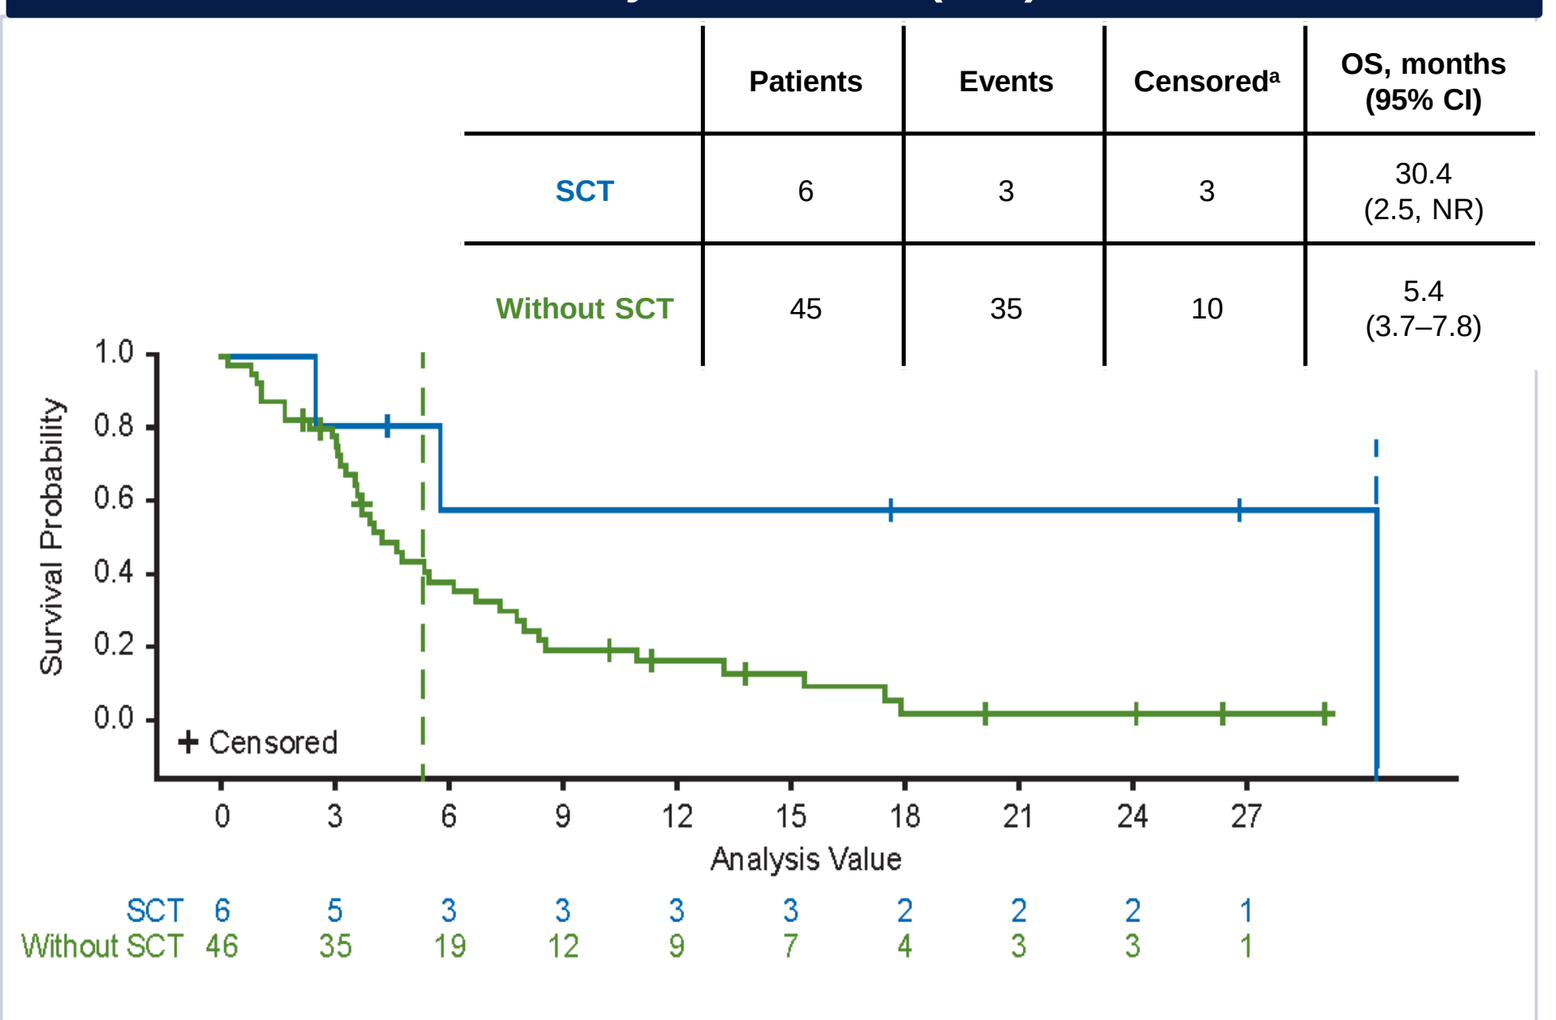
Patients
Events
Censored[a]
OS, months (95% CI)
SCT 6 3 3 30.4 (2.5, NR)
Without SCT 45 35 10 5.4 (3.7–7.8)
Survival Probability
Censored
Analysis Value
SCT 6 5 3 3 3 3 2 2 2 1
Without SCT 46 35 19 12 9 7 4 3 3 1

In the OS analysis, patients who were not confirmed to have died at time of analysis were censored on the last known alive date.
1L, frontline; CI, confidence interval; OS, overall survival; PCHM, prior or concomitant hematologic malignancy; PVEK, pivekimab sunirine; NR, not reached; R/R, relapsed or refractory; SCT, stem cell transplant.

Most common hematologic adverse events

Hematologic AEs[a] (≥15% of patients)

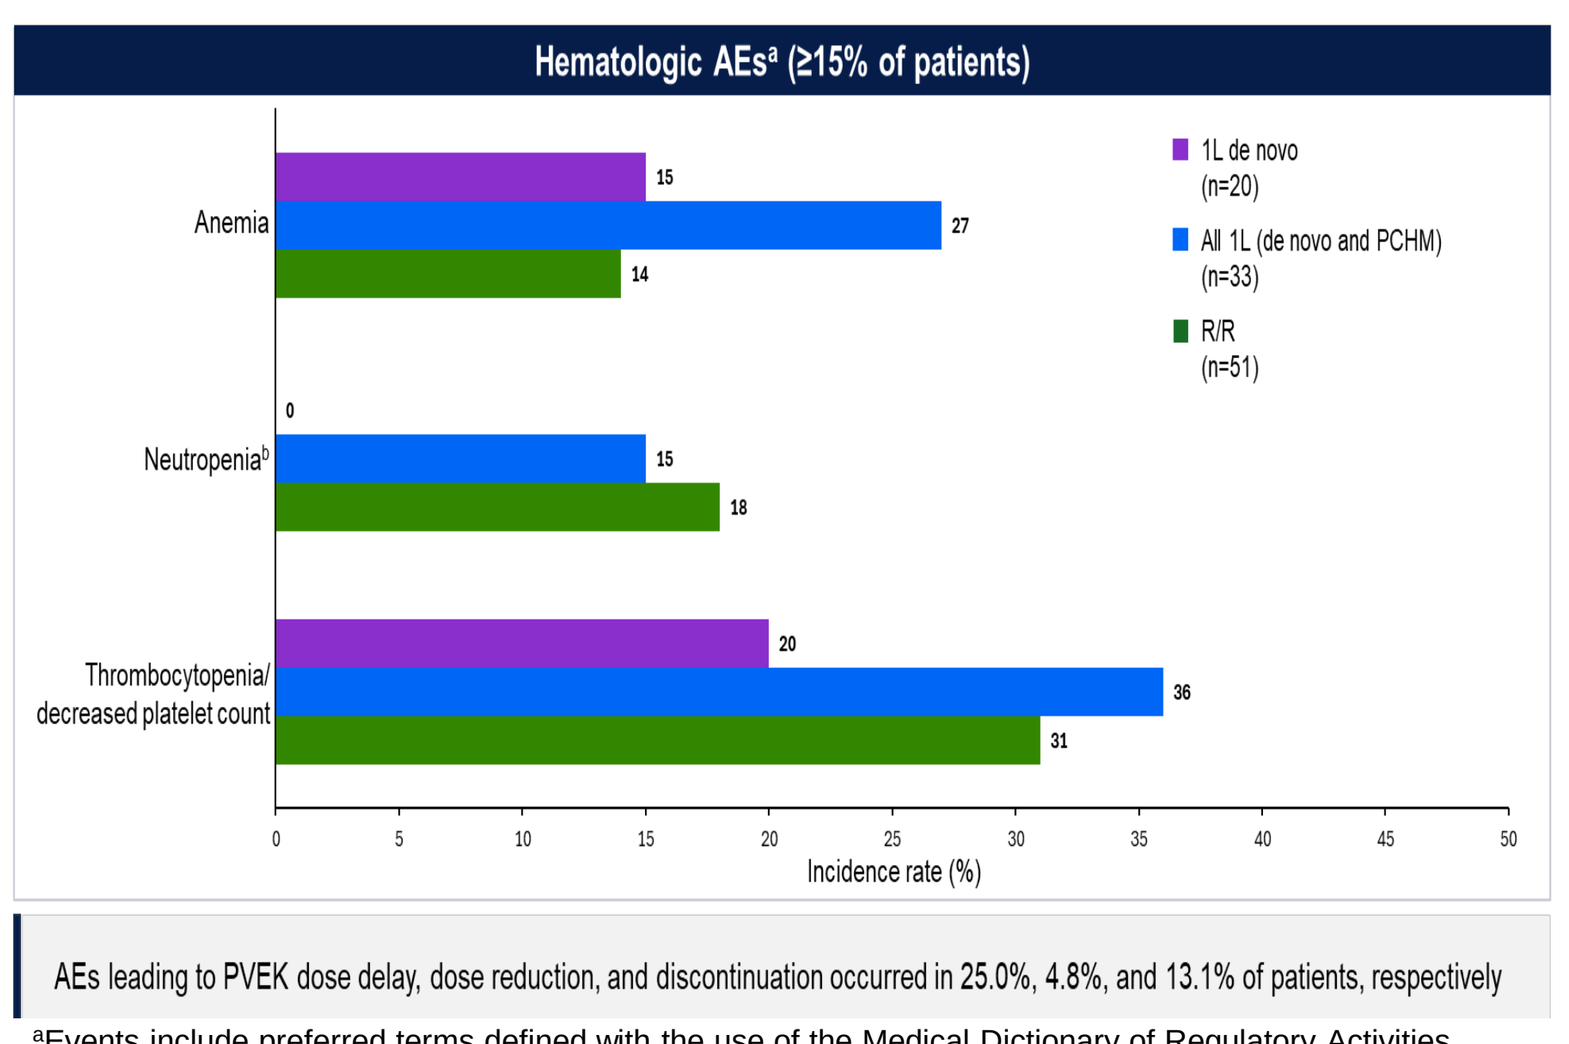
Anemia 15 27 14
Neutropenia[b] 0 15 18
Thrombocytopenia/ decreased platelet count 20 36 31
1L de novo (n=20)
All 1L (de novo and PCHM) (n=33)
R/R (n=51)
Incidence rate (%)

AEs leading to PVEK dose delay, dose reduction, and discontinuation occurred in 25.0%, 4.8%, and 13.1% of patients, respectively

[a]Events include preferred terms defined with the use of the Medical Dictionary of Regulatory Activities, version 27.1; [b]Granulocyte colony stimulating factor was administered to one patient in the frontline group with prior or concomitant hematologic malignancy. 1L, frontline; AE, adverse event; BPDCN, blastic plasmacytoid dendritic cell neoplasm; R/R, relapsed or refractory.

Non-hematologic AEs[a] (≥15% of patients[b])

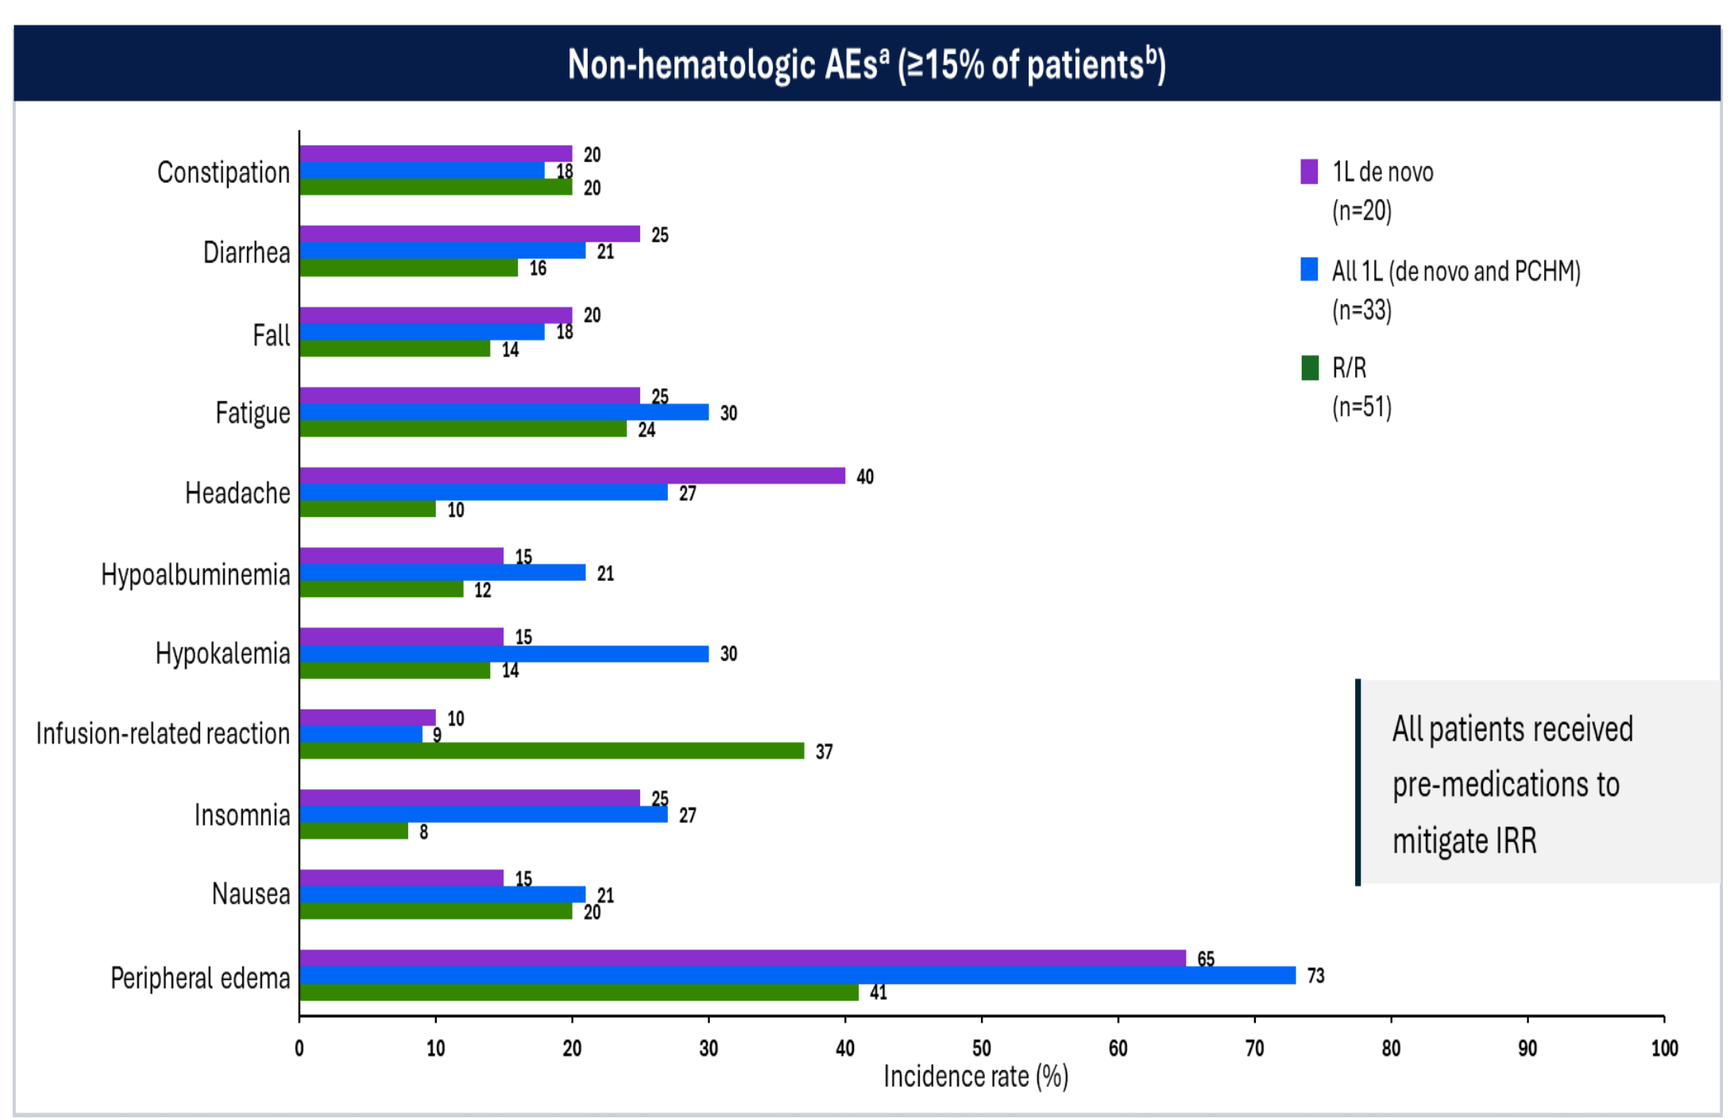
Constipation 20 18 20
Diarrhea 25 21 16
Fall 20 18 14
Fatigue 25 30 24
Headache 40 27 10
Hypoalbuminemia 15 21 12
Hypokalemia 15 30 14
Infusion-related reaction 10 9 37
Insomnia 25 27 8
Nausea 15 21 20
Peripheral edema 65 73 41
1L de novo (n=20)
All 1L (de novo and PCHM) (n=33)
R/R (n=51)
All patients received pre-medications to mitigate IRR
Incidence rate (%)

[a]Events include preferred terms defined with the use of the Medical Dictionary of Regulatory Activities, version 27.1; [b]Excludes AEs occurring at <15% in the total population (N=84). 1L, frontline; AE, adverse event; BPDCN, blastic plasmacytoid dendritic cell neoplasm; R/R, relapsed or refractory.

Individual serious and grade ≥3 AEs were generally low[a]

Serious AEs in ≥3% of patients

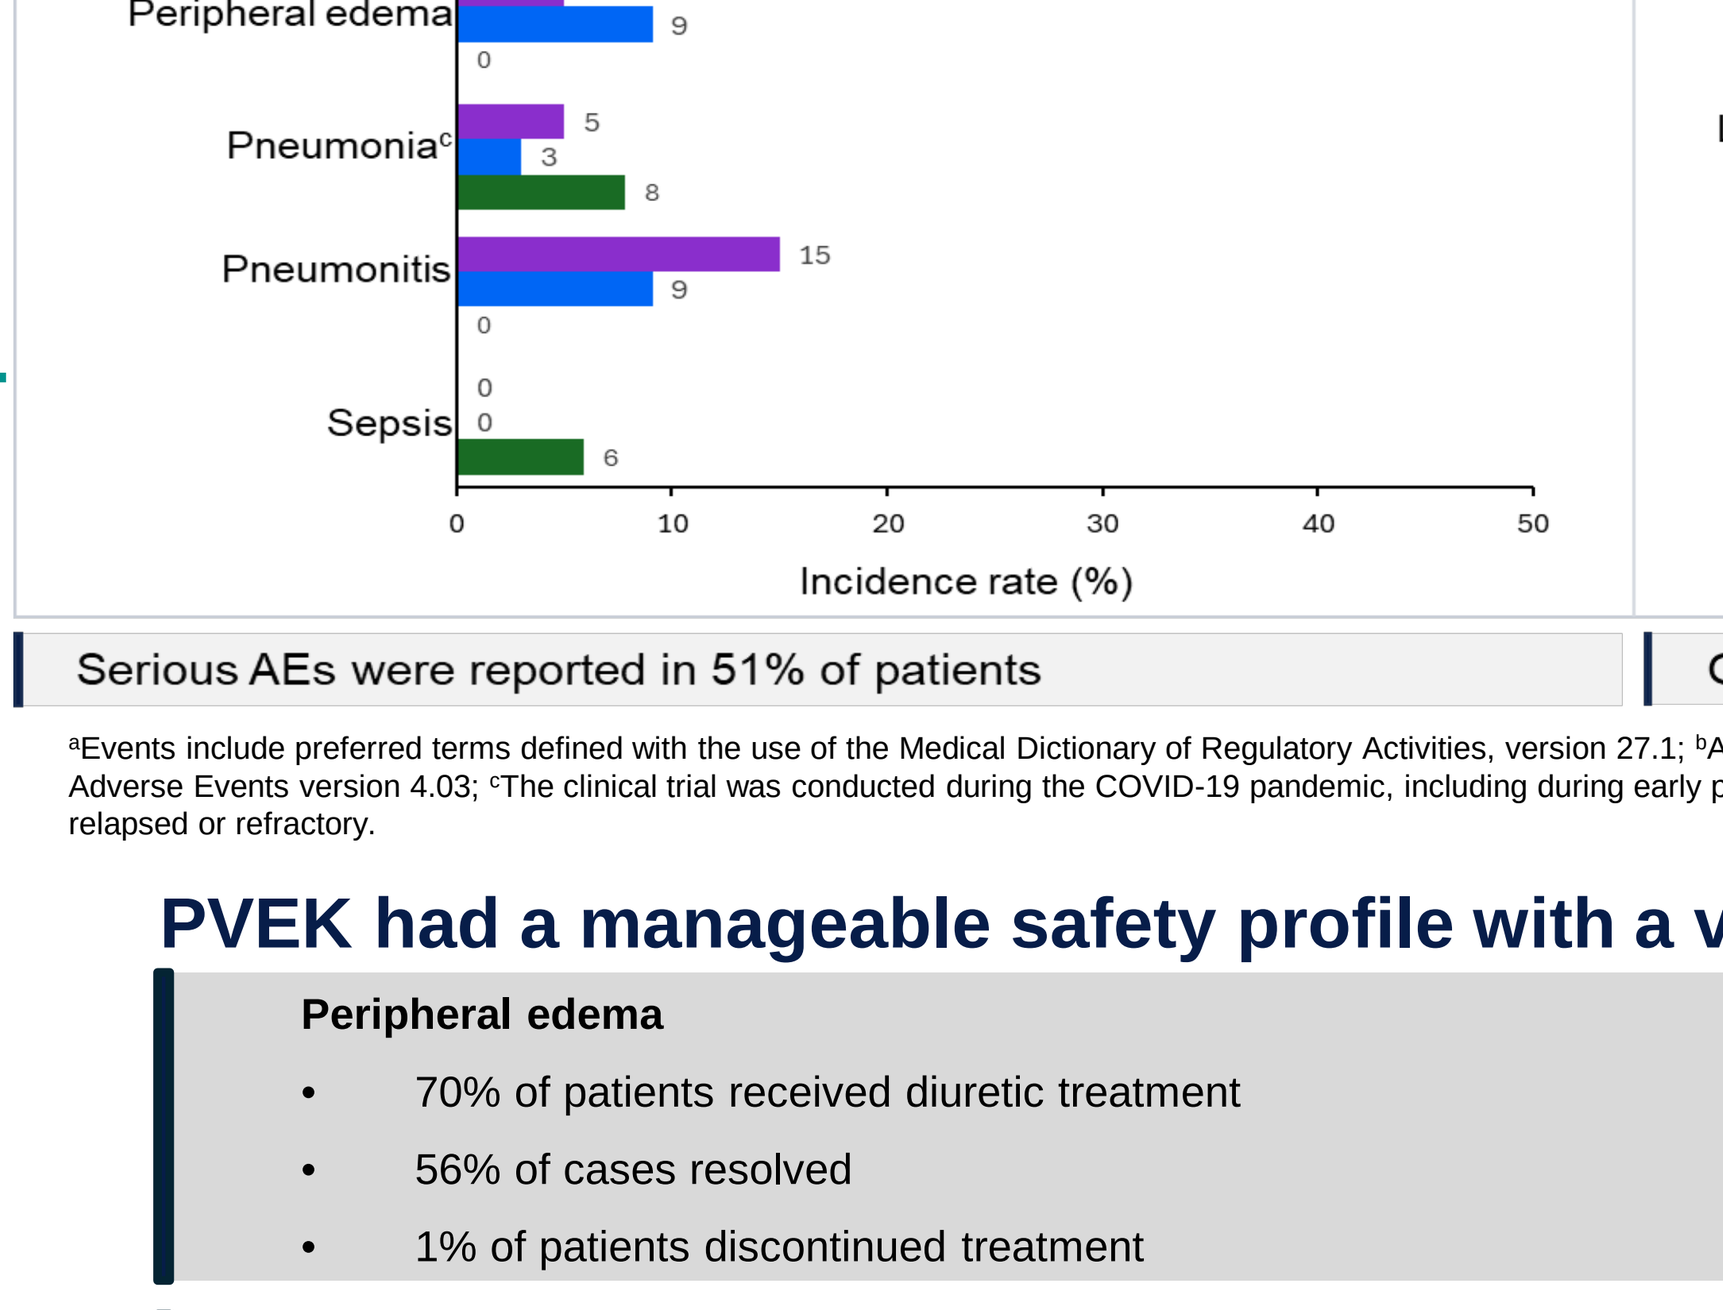
1L de novo (n=20)
All 1L (de novo and PCHM) (n=33)
R/R (n=51)
COVID-19[c] 10 6 2
Febrile neutropenia 5 3 6
Generalized edema 10 9 0
Peripheral edema 5 9 0
Pneumonia[c] 5 3 8
Pneumonitis 15 9 0
Sepsis 0 0 6
Incidence rate (%)

Grade ≥3 AEs in ≥10%[b]

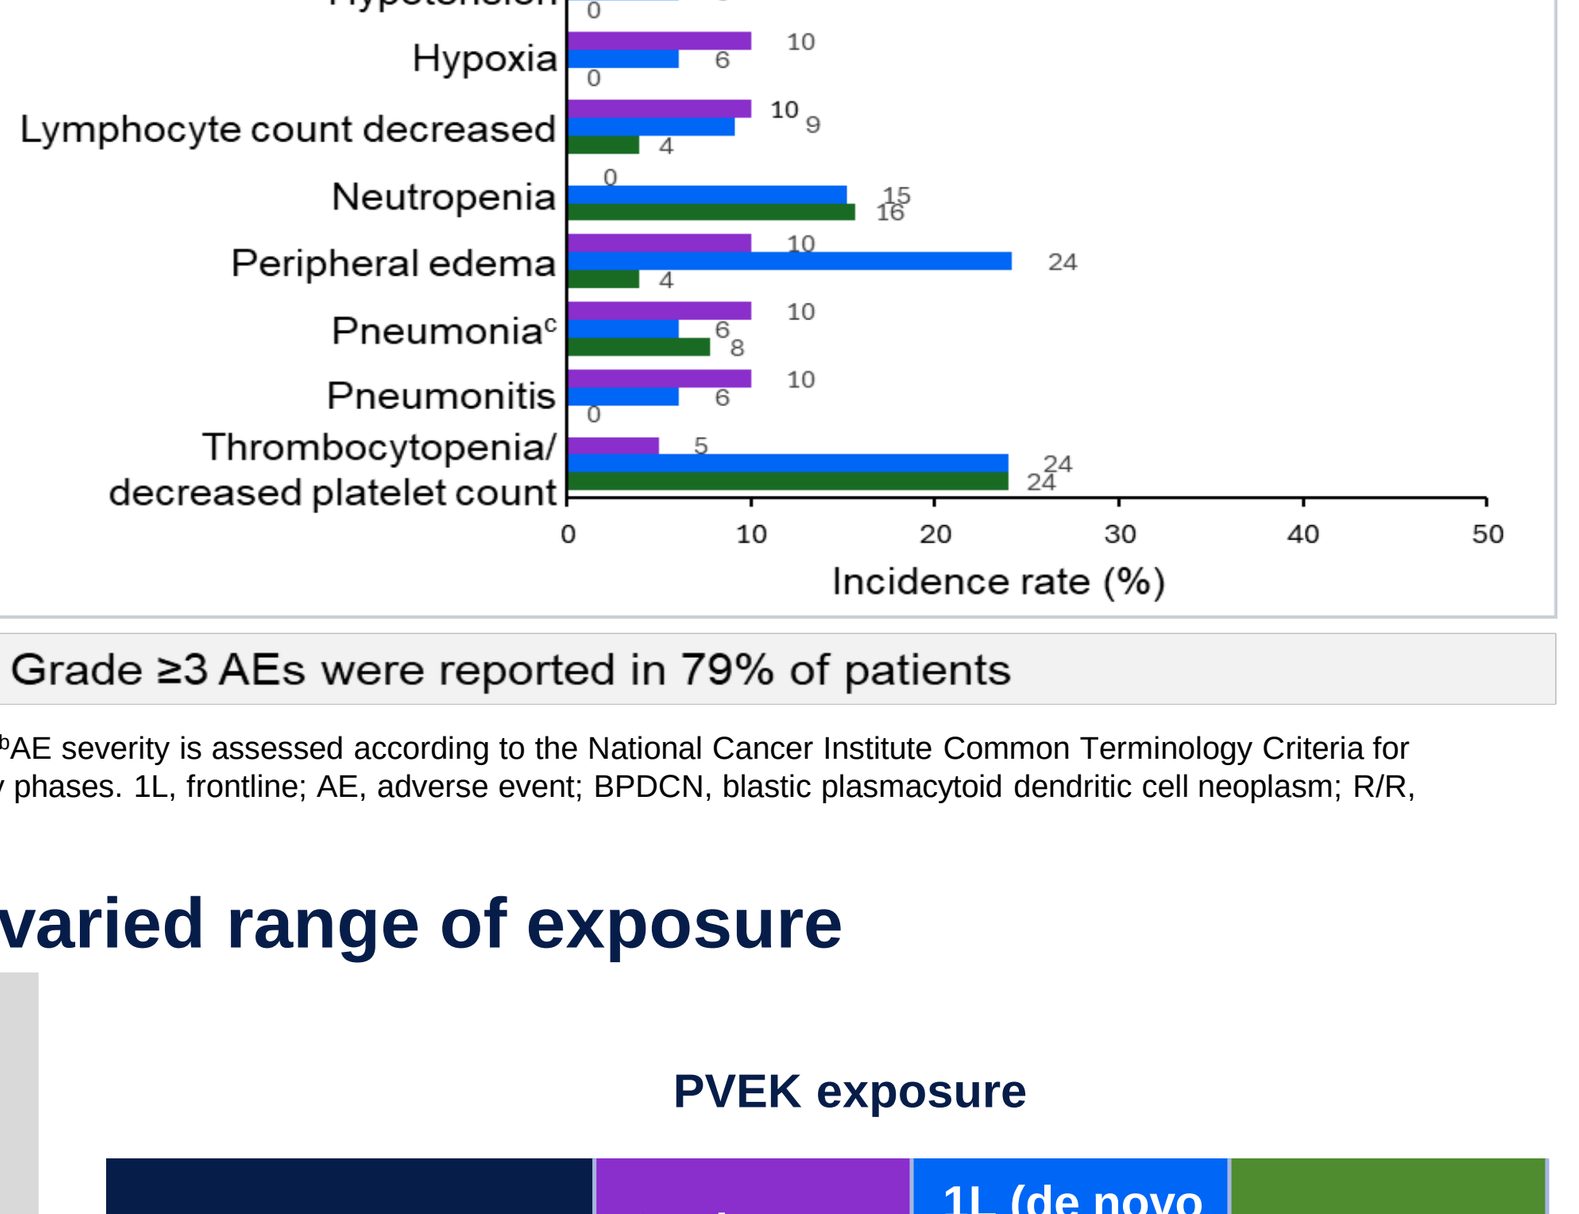
COVID-19[c] 10 6 2
Generalized edema 10 9 0
Headache 10 6 2
Hypertension 10 6 2
Hypophosphatemia 15 12 0
Hypotension 10 6 0
Hypoxia 10 6 0
Lymphocyte count decreased 10 9 4
Neutropenia 0 15 16
Peripheral edema 10 24 4
Pneumonia[c] 10 6 8
Pneumonitis 10 6 0
Thrombocytopenia/ decreased platelet count 5 24 24
Incidence rate (%)

Serious AEs were reported in 51% of patients

Grade ≥3 AEs were reported in 79% of patients

[a]Events include preferred terms defined with the use of the Medical Dictionary of Regulatory Activities, version 27.1; [b]AE severity is assessed according to the National Cancer Institute Common Terminology Criteria for Adverse Events version 4.03; [c]The clinical trial was conducted during the COVID-19 pandemic, including during early phases. 1L, frontline; AE, adverse event; BPDCN, blastic plasmacytoid dendritic cell neoplasm; R/R, relapsed or refractory.

PVEK had a manageable safety profile with a varied range of exposure

Peripheral edema
- 70% of patients received diuretic treatment
- 56% of cases resolved
- 1% of patients discontinued treatment

Infusion-related reactions
- Did not lead to treatment discontinuation
- Mild to moderate in severity (only 4% at maximum grade 3)
- Short duration (resolving in median of 1 [1–22] day)
- Preventable with corticosteroids

Two on-treatment **veno-occlusive disease**
- Resolved but led to treatment discontinuation

Zero **capillary-leak syndrome** events

PVEK exposure

	1L de novo BPDCN (n=20)	1L (de novo and PCHM) BPDCN (n=33)	R/R BPDCN (n=51)
Median duration of PVEK exposure, weeks (range)	15.4 (6.3–86.0)	16.4 (6.3–86.0)	9.0 (1.0–128.9)
Median number of PVEK cycles (range)	5 (2–23)	5 (2–23)	3 (1–34)

1L frontline; AE, adverse event; BPDCN, blastic plasmacytoid dendritic cell neoplasm; PCHM, prior or concomitant hematologic malignancy; PVEK, pivekimab sunirine; R/R, relapsed or refractory.

Scan QR code to download an electronic version of this presentation: Previously presented at EHA 2025
QR Code expiration: August 1, 2026
To submit a medical question, please visit www.abbviemedinfo.com
DV-016232

References:
1. Pemmaraju N, et al. *Blood*. 2023;141(6):567–578;
2. Pemmaraju N, et al. *N Engl J Med*. 2019;380(17):1628–1637;
3. Cuglievan B, et al. *Leukemia*. 2023;37:1767–1778;
4. Pemmaraju N, et al. *HemaSphere*. 2023;7(S3).
5. Kovtun Y, et al. *Blood Adv*. 2018;2(8):848–858.

Acknowledgments

AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the abstract. All authors had access to relevant data and participated in the drafting, review, and approval of this abstract. No honoraria or payments were made for authorship. Medical writing support was provided by Sean Hourihane, PhD, and Brandy Menges, PhD, of Fishawack Facilitate Ltd, part of Avalere Health and was funded by AbbVie.

Disclosures

Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: **NP:** Consultant or advisor for Blueprint Medicines, Pacylex, Immunogen, Bristol-Myers Squibb, Clearview Healthcare Partners, Astellas Pharma, CTI BioPharma Corporation, AbbVie Inc, Aptitude Health, Aplastic Anemia and MDS International Federation, CancerNet, CareDx, Celgene, Cimeio Therapeutics, Curio Science, Dava Oncology, EUSA Pharma, Harborside Press, Imedex, Intellisphere, Magdalen Medical Publishing, Medscape, Menarini Group, Neopharm, Novartis, OncLive, Patient Power, Peerview, PharmaEssentia, Physician's Education Resource; Leadership roles in ASH; Other relationships with Karger Publishers; Institutional funding from US Department of Defense. **GM:** Honoraria, consulting or advisor from AbbVie Inc, Astellas Pharma, AstraZeneca, ImmunoGen, Janssen, Menarini Group, Pfizer, Ryvu Therapeutics, SERVIER, Syros Pharmaceuticals; Speakers' bureau: AbbVie Inc, Astellas Pharma, AstraZeneca, ImmunoGen, Janssen, Menarini Group, Pfizer, Ryvu Therapeutics, Syros Pharmaceuticals, Takeda; Research funding from AbbVie Inc, Pfizer, Jazz Pharmaceuticals, AstraZeneca, MEI Pharma, and Daiichi Sankyo; Travel expenses from Kura Oncology and Jazz Pharmaceuticals. **PM:** Consultant or advisor for AbbVie Inc, Pfizer, Daiichi Sankyo, Jazz Pharmaceuticals, SERVIER, Otsuka; Speakers' bureau: Otsuka, Celgene, Daiichi Sankyo; Travel expenses from Amgen; institutional research funding from Celgene, Janssen-Cilag, Pfizer, SERVIER, AbbVie Inc, Daiichi Sankyo Europe GmbH. **AAL:** Stock or ownership interests in Stelexis BioSciences Inc; consultant or advisor for Cimeio Therapeutics, IDRx, Jnana Therapeutics, ProteinQure, Qiagen; research funding from AbbVie Inc, Stemline Therapeutics. **LM:** Nothing to disclose. **DAS:** Consultant or advisor for AbbVie Inc, Bristol-Myers Squibb, Gilead Sciences, Kite (a Gilead company), Novartis, Syndax, Janssen, SERVIER, Syros Pharmaceuticals, Takeda, Molecular Partners, AvenCell, Agios, Dark Blue Therapeutics; researching funding from Jazz Pharmaceuticals and Aprea Therapeutics (institution). **MLU:** Consultant or advisor for ADC Therapeutics, Bristol-Myers Squibb/Celgene/Juno, Autolus, Genentech, Stemline Therapeutics, Kura Oncology; travel expenses from Miltenyi Biotec; research funding from Kite (a Gilead Company) and Immunogen. **GJS:** Consultant or advisor for Ono Pharmaceuticals, Agios, Celgene, Incyte, Jazz Pharmaceuticals, Novartis, AbbVie Inc, Astellas Pharma, Autolus Therapeutics, Rigel, Bristol-Myers Squibb/Celgene; speakers' bureau for Astellas Pharma, Kite (a Gilead Company), Jazz Pharmaceuticals, Stemline Therapeutics, Bristol-Myers Squibb, Sanofi, Karyopharm Therapeutics, Incyte, AbbVie Inc, Rigel, Blueprint Medicines, Agios, Seattle Genetics/Astellas, Amgen; stock and other ownership interests in Bristol-Myers Squibb, Amgen, Johnson and Johnson; researching funding from AbbVie Inc, Actinium Pharmaceuticals, Actuate Therapeutics, Arog, Astellas Pharma, Bristol-Myers Squibb/Celgene, Celator, Constellation Pharmaceuticals, Daiichi Sankyo, Deciphera, Delta-Fly Pharma, FORMA Therapeutics, Fujifilm, Gamida Cell, Genentech Roche, Geron, Incyte, Karyopharm Therapeutics, Kite (a Gilead Company), Mateon Therapeutics, Onconova Therapeutics, Pfizer, PrECOG, REGIMMUNE, Samus Therapeutics, Sangamo Bioscience, Stellas Life Sciences, Stemline Therapeutics, Takeda, Tolero Pharmaceuticals, Trovagene, Agios, Amgen, Jazz Pharmaceuticals, ElevateBio, Ono Pharmaceutical, Novartis, Sanofi, AVM Biotechnology, Syros Pharmaceuticals, GlycoMimetics, Aptevo Therapeutics, Biomea Fusion, Bio-Path Holdings, Biosight, Cellectis, Celularity, Cogent Biosciences, Cullinan Oncology, Fate Therapeutics, Immune-Onc Therapeutics, Kronos Bio, Kura Oncology, Loxo, Marker Therapeutics. **HPE:** Consultant or advisor for Agios, Astellas Pharma, Amgen, Celgene, Daiichi Sankyo, GlycoMimetics, Immunogen, Inctye, Jazz Pharmaceuticals, Marcrogenics, Novartis, AbbVie, Genentech Roche, Janssen Oncology, Pfizer, Trillium Therapeutics, Takeda, Kura Oncology, Bristol-Myers Squibb/Celgene, Sumitomo Dainippon Pharma Oncology, Schrodinger, Stemline Therapeutics, SERVIER; Speakers' bureau for Agios, Incyte, Jazz Pharmaceuticals, Novartis, Bristol-Myers Squibb Foundation, AbbVie Inc, SERVIER, Syndax; other relationships – GlycoMimetics, Celgene, Bristol-Myers Squibb/Celgene; institutional researching funding from AbbVie, Agios, Amgen, Daiichi Sankyo, Forma Therapeutics, Gilead/Forty Seven, Immunogen, Jazz Pharmaceuticals, Macrogenics, Novartis, PTC Therapeutics, AbbVie Inc, GlycoMimetics, ALX Oncology, SERVIER, Aptose Biosciences, Sumitomo Dainippon Pharma Oncology, Ascentage Pharma, Kura Oncology, Oryzon Genomics, Taiho Oncology. **ESW:** Consultant or advisor for AbbVie Inc, Kite/Gilead, Bristol-Myers Squibb/Celgene, Novartis, Kura Oncology, CTI BioPharma Corp, Daiichi Sankyo/UCB Japan, Rigel, Sumitomo Pharma Oncology, Immunogen, Schrodinger, Qiagen, Syndax, Blueprint Medicines, Johnson and Johnson/Janssen, SERVIER, Takeda, Stemline Therapeutics, Ryvu Therapeutics, GlaxoSmithKline; speakers' bureau for Pfizer, Astellas Pharma, DAVA Pharmaceuticals, Daiichi Sankyo/UCB Japan; other relationships – AbbVie Inc, Genentech Roche, Gilead Sciences, UpToDate. **RBW:** Consultant or advisor for Eigen, Omeros, and Wugen; researching funding from ImmunoGen/AbbVie and Pfizer and institutional research funding from Jazz Pharmaceuticals, ImmunoGen/AbbVie, Kura Oncology, and Vor BioPharma. **ED:** Consultant or advisor for Stemline Therapeutics; institutional research funding from ImmunoGen, Roche, Novartis, Pfizer, AbbVie Inc, Janssen; travel expenses from Stemline Therapeutics, ImmunoGen, Novartis, Pfizer, Janssen-Cilag, Grifols. **AMA**: Consultant or advisor for Seagen. **OL:** Nothing to disclose. **DL**: Nothing to disclose. **GM:** Nothing to disclose. **DJD**: Consultant or advisor for Pfizer, Amgen, Novartis, Takeda, Blueprint Medicines, Incyte, Gilead Sciences, Jazz Pharmaceuticals, SERVIER; institutional research funding from Novartis, AbbVie Inc, GlycoMimetics, and Blueprint Medicines. **YD:** Employee of ImmunoGen (now AbbVie Inc) and owns AbbVie stocks. **JP**: Employee of AbbVie Inc. and may own stock or stock options. **NGD**: Consultant or advisor for Celgene, Agios, Jazz Pharmaceuticals, Pfizer, AbbVie Inc, Astellas Pharma, Daiichi Sankyo, Novartis, Bristol-Myers Squibb, Amgen, Immunogen, Genentech, SERVIER, Syndax, Trillium Therapeutics, Gilead Sciences, Arog, Shattuck Labs, Kite (a Gilead Company), Menarini, Rigel; institutional research funding from Bristol-Myers Squibb, Pfizer, Immunogen, Genentech, AbbVie Inc, Astellas Pharma, SERVIER, Daiichi Sankyo, Gilead Sciences, Amgen, Trillium Therapeutics, Hanmi, Trovagene, FATE Therapeutics, Novimmune, GlycoMimetics, Kite (a Gilead Company).

Presented at the Society of Hematologic Oncology (SOHO) Annual Meeting; September 3–6, 2025; Houston, TX