MDS-1497

Primary Analysis of the Randomized, Phase 3 VERONA Study of Venetoclax With Azacitidine Versus Placebo With Azacitidine in Patients With Treatment-Naïve, Intermediate and Higher-Risk Myelodysplastic Syndromes

Guillermo Garcia-Manero, MD[1]; Uwe Platzbecker, MD[2]; Pierre Fenaux, MD, PhD[3]; Gail J. Roboz, MD[4]; Chun Y. Fong, PhD, FRACP, FRCPA, MBBS[5]; Marek Hus, MD, PhD, DSc[6]; Robert Delage, MD, MSc, FRCP[7]; Andrew M. Brunner, MD[8]; Je-Hwan Lee, MD, PhD[9]; Mehmet Turgut, MD, PhD[10]; Dominiek Mazure, MD[11]; Yin-Hsun Feng, MD, PhD[12]; Yasushi Miyazaki, MD[13]; Zdeněk Kořístek, MD, PhD[14]; Sophie Dimicoli-Salazar, MD, PhD[15]; María D. Campelo, MD, PhD[16]; David Lavie, MD[17]; Dominic Culligan, MD[18]; Grace Ku, MD[19]; Zhijian Xiao, MD, PhD[20]; Fang Fang, PhD[21]; Maggie Headley, MD[21]; Jalaja Potluri, MD, FACP[21]; Amer M. Zeidan, MBBS, MHS[22]*; Jacqueline Garcia, MD[23]*

*Authors Contributed Equally

[1]University of Texas MD Anderson Cancer Center, Houston, TX USA; [2]Universitätsklinikum Leipzig, Saxony, Germany; [3]Hôpital St. Louis/Assistance Publique- Hôpitaux de Paris and Université de Paris, Paris, France; [4]Weill Cornell Medical College, New York, NY, USA; [5]Austin Health, Melbourne, Australia; [6]Medical University of Lublin, Lublin, Poland; [7]Hôpital de l'Enfant-Jésus, Quebec City, Quebec, Canada; [8]Massachusetts General Hospital, Boston, MA, USA; [9]University of Ulsan College of Medicine, Seoul, Korea; [10]Ondokuzmayıs University Medical Faculty, Samsun, Turkey; [11]Universitair Ziekenhuis Gent, Gent, Belgium; [12]Chi-Mei Medical Center, TAIWAN (PROVINCE OF CHINA); [13]Atomic Bomb Disease Institute, Nagasaki University, Nagasaki, Japan; [14]University Hospital Ostrava, Ostrava, Czech Republic; [15]CHU Bordeaux - Hôpital Haut Lévêque, Pessac, France; [16]Hospital Universitario de Salamanca, IBSAL, Universidad de Salamanca, Salamanca, Spain; [17]Hadassah Medical Center, Jerusalem, Israel; [18]Aberdeen Royal Infirmary, Aberdeen, United Kingdom; [19]Genentech Inc., South San Francisco, CA, USA; [20]Chinese Academy of Medical Sciences and Peking Union Medical College, Tianjin, China; [21]AbbVie Inc., North Chicago, IL, USA; [22]Yale School of Medicine, Yale University, New Haven, CT, USA; [23]Dana-Farber Cancer Institute, Boston, MA, USA

OBJECTIVE

To evaluate the efficacy and safety of venetoclax in combination with azacitidine for the treatment of patients with treatment-naïve intermediate and higher-risk MDS

CONCLUSIONS

Ven + Aza did not show an improvement in OS (primary endpoint) compared to Pbo + Aza in treatment-naïve patients with IPSS-R intermediate and higher-risk MDS

Ven + Aza resulted in a higher mOR vs Pbo + Aza in this patient population

The safety risks of Ven + Aza were as expected and manageable; no new safety signals were observed

AbbVie and the authors thank the participants, study sites, and investigators who participated in this clinical trial.

AbbVie and Genentech/Roche funded this trial and participated in the trial design, research, analysis, data collection, interpretation of data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. The authors thank Marianne Pons, PhD, and Phillip Giannopoulos, PhD, CMPP, of Bio Connections, LLC, for medical writing support funded by AbbVie.

Financial arrangements of the authors with companies whose products may be related to the present report are listed as declared by the authors: KI reports advisory board and honoraria fees from AbbVie; **GGM:** research support from AbbVie and Genentech; **UP**: member of Board of Directors or advisory committee for Bristol Myers Squibb and MDS Foundation; honoraria, consultancy, and research funding from Bristol Myers Squibb, Novartis, Jazz, Syros, Servier, Silence Therapeutics, and Takeda; consultancy and research funding from Curis, Geron, and Janssen Biotech; research funding from BMS, BeiGene, FibroGen, Roche, and Merk; consultancy from AbbVie; and honoraria from Celgene; **PF**: consultancy/advisory role for Bristol Myers Squibb, AbbVie, Janssen, Jazz, and Novartis; research support, as GFM chair, from Bristol Myers Squibb, AbbVie, Janssen, Jazz, and Novartis; honoraria from Bristol Myers Squibb, AbbVie, Janssen, Jazz, and Novartis; **GJR**: consultant for AbbVie, Amgen, AstraZeneca, Bristol Myers Squibb, Caribou Biosciences, Celgene, Daiichi Sankyo, Ellipses Pharma, Genoptix, Geron, GlaxoSmithKline, Janssen, Jasper Pharmaceuticals, Laxx Pharmaceuticals, Molecular Partners, Novartis, Pfizer, Oncoverity, OncoPrecision, Rigel, Roche, Syndax, Takeda (IRC Chair), and Telix Pharma; research support from Janssen; **CYF**: honoraria from AbbVie, Amgen, BeiGene, Bristol Myers Squibb, Jazz, Novartis, Pfizer, and Servier; consultant/advisor for AbbVie, Amgen, Astellas Pharma, Bristol Myers Squibb, Jazz, Novartis, Pfizer, and Servier; research funding from Amgen, Astellas, and Jazz; **YM**: honoraria from Novartis, Celgene, Sumitomo Pharma, Nippon Shinyaku, Chugai, Otsuka, Astellas, and Kyowa-Kirin; research funding from Chugai, and Sumitomo Pharma; **MDC**: honoraria/speaker from BMS, Novartis, Keros, and AbbVie; advisory boards for BMS, Novartis, GSK, Agios, Hemavant, Syros, Keros, Curis, Astex/Otsuka, Ascentage, Fortrea, AstraZeneca; **DL**: honoraria for lectures or advisory boards: AbbVie, Medison, MSD, Novartis, Roche, Takeda; **DC**: advisory boards from AbbVie, BMS, Jazz and Roche, and pharmaceutical meeting support for Jazz and Servier; **GK**: employment with Genentech; may hold stock or other options from Roche; **FF, MH, JP**: employment with AbbVie; may hold stock or other options; **AZ**: advisory boards, consulted, participated in clinical trial committees, and/or received honoraria from AbbVie, Akesobio, Agios, Amgen, Astellas, BioCryst, Beigene, Boehringer-Ingelheim, Celgene/BMS, Chiesi/Cornerstone biopharma, Daiichi Sankyo, Dr Reddy, Epizyme, Faron, Fibrogen, GSK, Glycomimetics, Genentech, Gilead, Geron, Janssen, Jasper, Karyopharm, Kyowa Kirin, Keros, Kura, Novartis, Notable, Orum, Otsuka, Pfizer, Regeneron, Rigel, Seattle Genetics, Shattuck labs, Schrodinger, Syros, Syndax, Servier, Takeda, Treadwell, Taiho, Vincerx, and Zentalis; **JG**: advisory role for AbbVie, Astellas, Bristol Myers Squibb, Genentech, and Servier; institutional funding from AbbVie, AstraZeneca, Genentech, New Wave, Prelude, and Pfizer; **MH, RD, AB, J-HL, MT, DM, Y-HF, ZK, SD-S, ZX**: Nothing to disclose.

References

1. Kröger, N. *Hematologica*. 2025;110(2):339-349.
2. Garcia, JS et al. *Blood*. 2025;145:1126-1135.

Abbreviations

AE, adverse event; AML, acute myeloid leukemia; ASCT, allogenic stem cell transplantation; Aza, azacitidine; BCL-2, B-cell lymphoma 2; CI, confidence interval; CR, complete remission; ECOG PS, Eastern Cooperative Oncology Group Performance Status; HMA, hypomethylating agent; HI, hematological improvement; HR, hazard ratio; HR MDS higher risk myelodysplastic syndromes; HSCT, hematopoietic stem cell transplantation; IPSS-R, Revised International Prognostic Scoring System; IV, intravenous; IVRS, Interactive Voice Response System; IWG, International Working Group; mCR, marrow complete remission; MDS, myelodysplastic syndromes; mOR, modified overall response; OR, overall response; OS, overall survival; Pbo, placebo; PD, progressive disease; PO, oral; PR, partial remission; PRO, patient-reported outcome; QD, once daily; SC, sub-cutaneous; TEAE, treatment-emergent adverse event; Ven, venetoclax; WHO, World Health Organization.

For additional information or to obtain a PDF of this poster

Scan QR code to download an electronic version of this presentation and other AbbVie SOHO 2025 scientific presentations:

QR code expiration: August 06, 2026

To submit a medical question, please visit www.abbviemedinfo.com

INTRODUCTION

- Intermediate, high and very high risk[a] (HR) MDS is an aggressive disease with a median overall survival of 3 years and 1.6 to 0.8 years, respectively with HMA alone[1]
- The only potentially curative treatment is allogeneic stem cell transplantation, but most patients are ineligible, representing a significant unmet need[1]
- In a Phase 1b study in treatment-naïve patients with HR MDS, the selective, BCL-2 inhibitor venetoclax in combination with the HMA azacitidine had a modified overall response (complete remission + partial remission + marrow complete remission) rate of 80.4%[2]
 - The venetoclax and azacitidine combination had manageable safety profile[2]

[a]IPSS-R score >3.

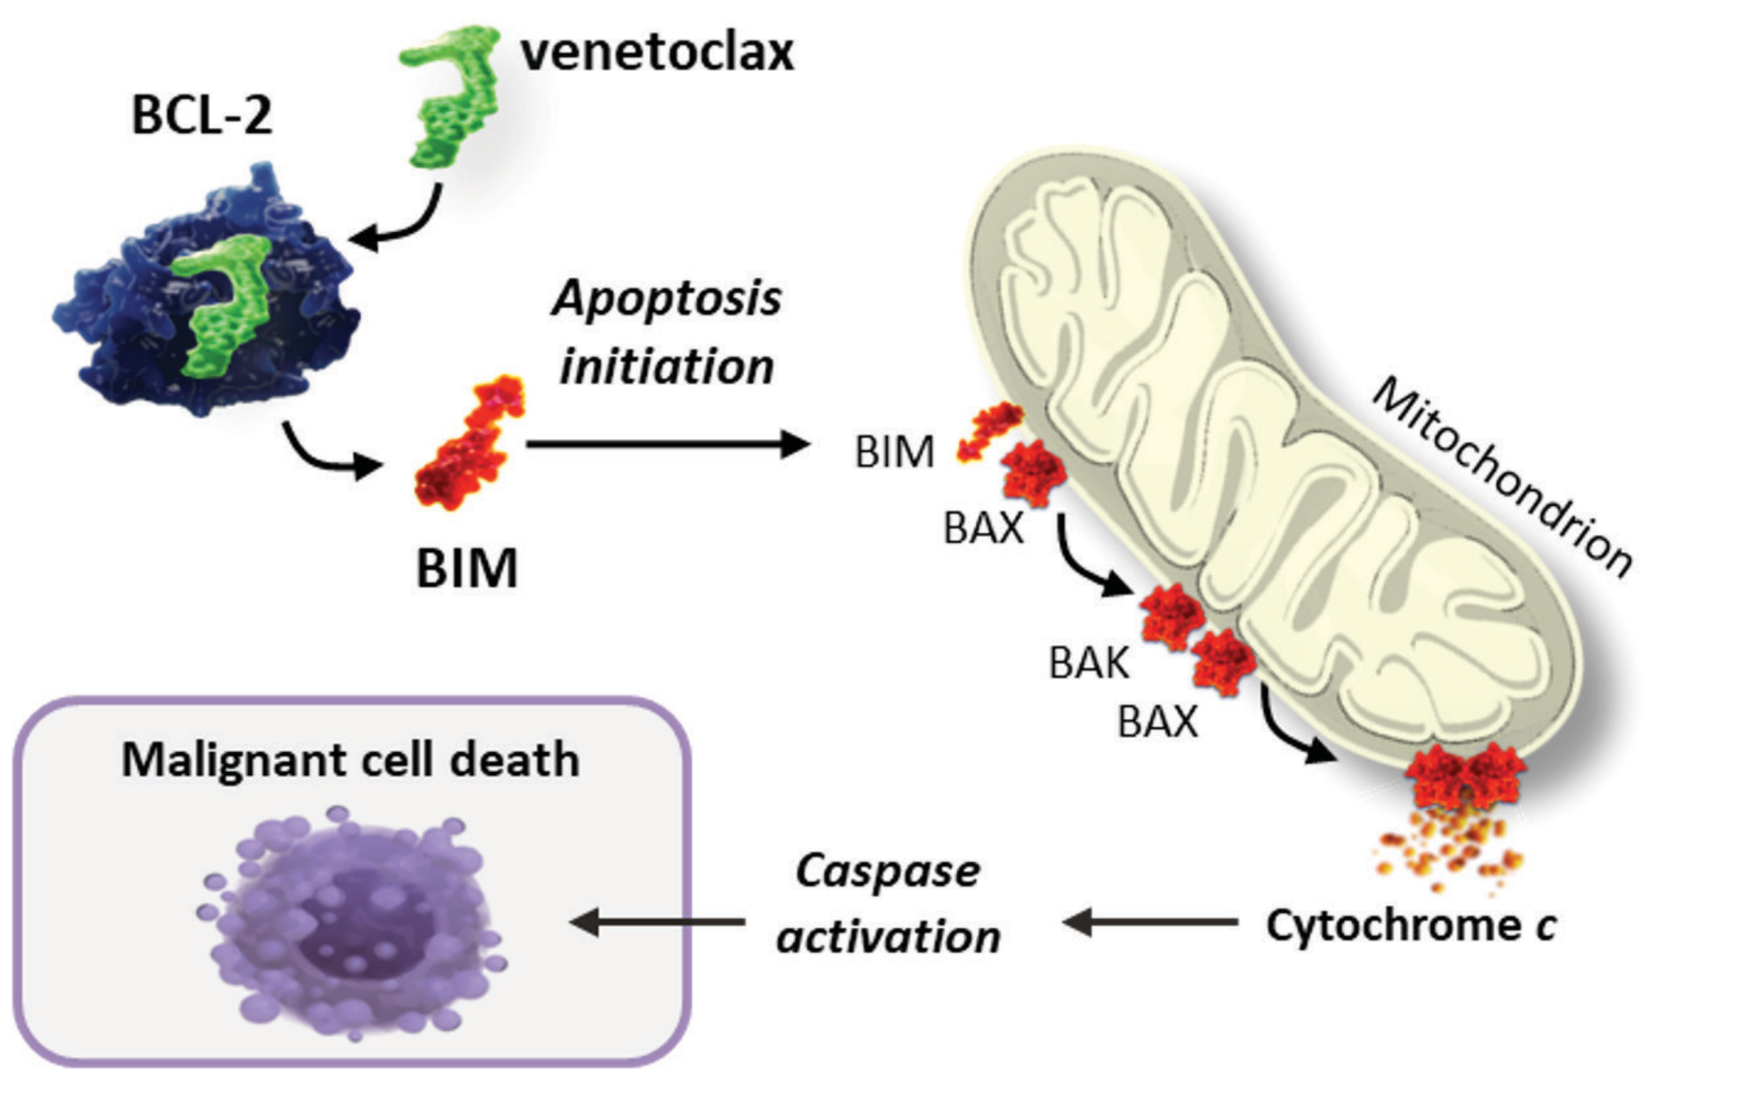
BCL-2
venetoclax
Apoptosis initiation
BIM
BIM
BAX
Mitochondrion
BAK
BAX
Malignant cell death
Caspase activation
Cytochrome c

We report efficacy and safety of venetoclax in combination with azacitidine from the primary analysis of the Phase 3 VERONA trial (NCT04401748)

METHODS

Figure 1. Study Design

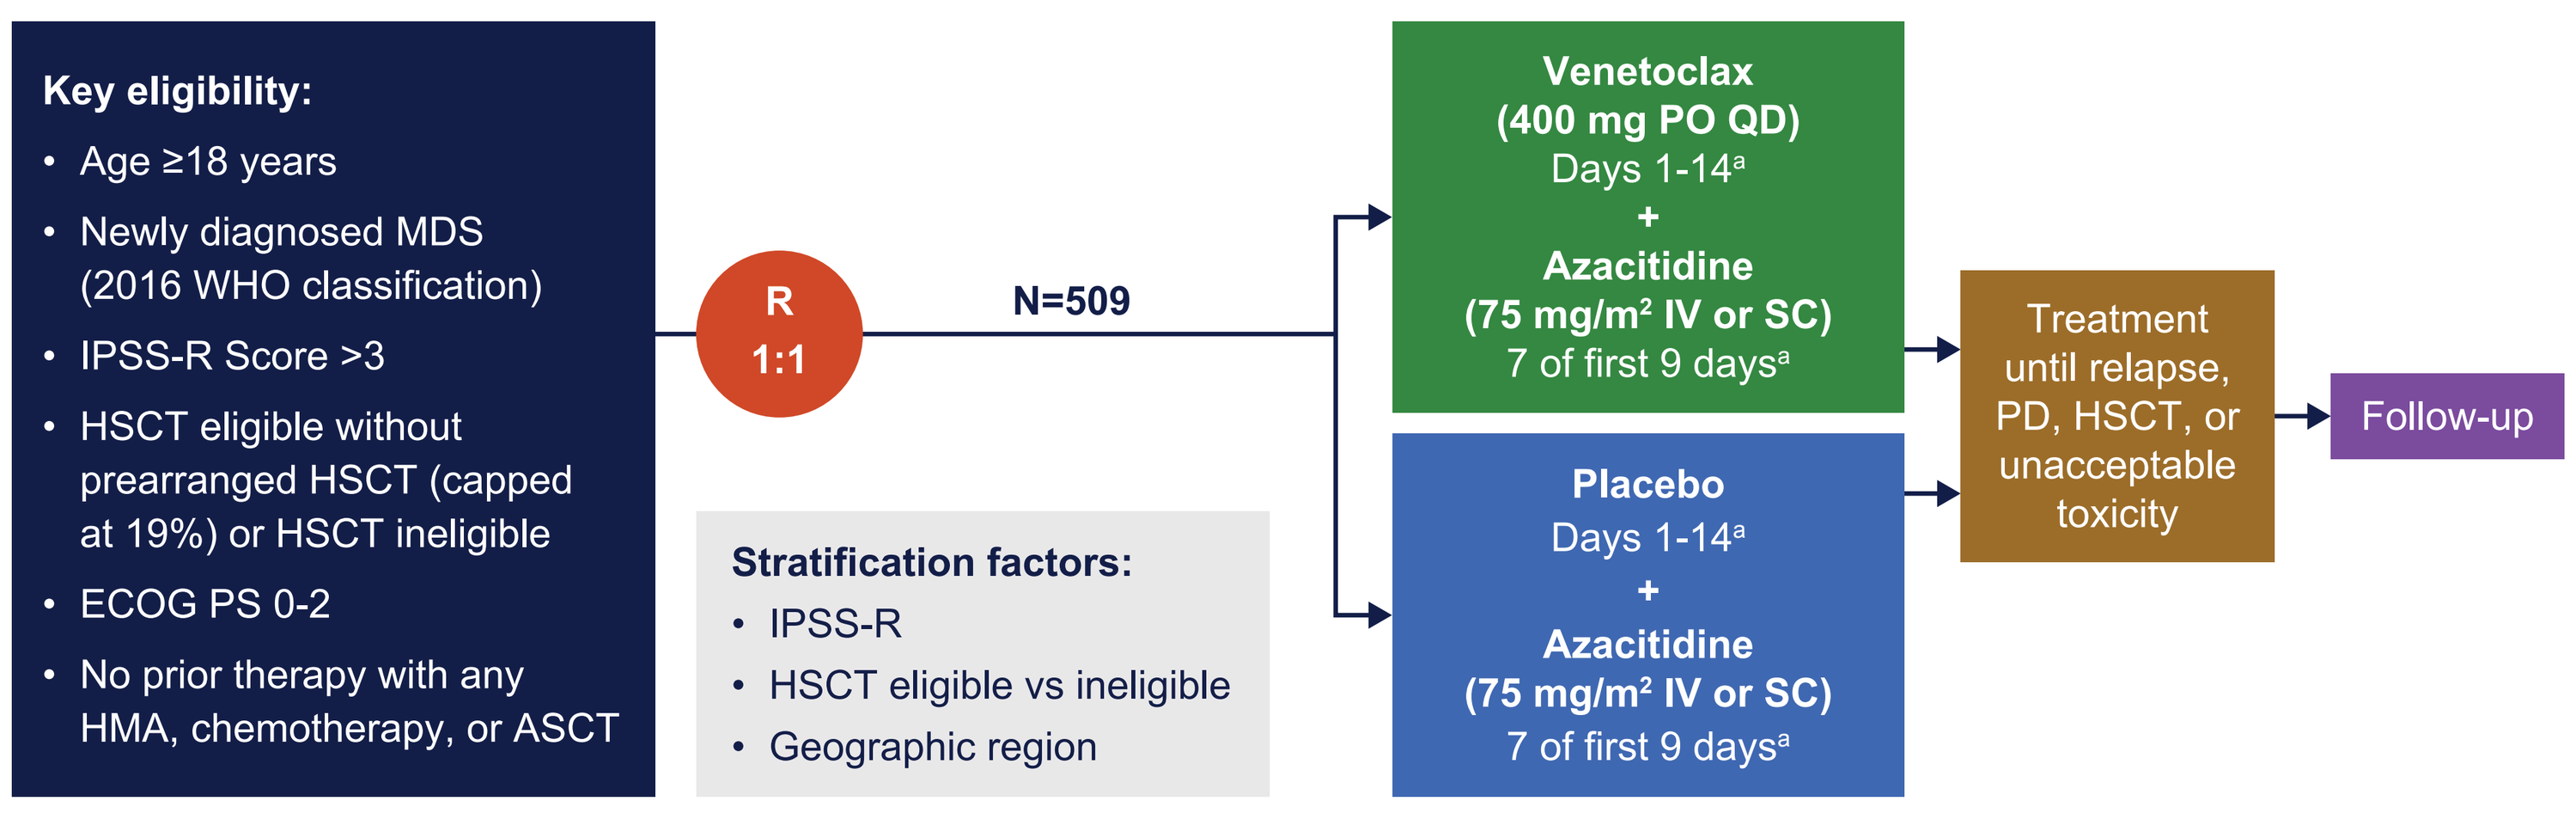
Key eligibility:
Age ≥18 years
Newly diagnosed MDS (2016 WHO classification)
IPSS-R Score >3
HSCT eligible without prearranged HSCT (capped at 19%) or HSCT ineligible
ECOG PS 0-2
No prior therapy with any HMA, chemotherapy, or ASCT
R 1:1
N=509
Stratification factors:
IPSS-R
HSCT eligible vs ineligible
Geographic region
Venetoclax (400 mg PO QD) Days 1-14[a] + Azacitidine (75 mg/m² IV or SC) 7 of first 9 days[a]
Placebo Days 1-14[a] + Azacitidine (75 mg/m² IV or SC) 7 of first 9 days[a]
Treatment until relapse, PD, HSCT, or unacceptable toxicity
Follow-up

- VERONA (NCT04401748) is a Phase 3 trial evaluating the efficacy and safety of BCL-2 inhibitor venetoclax in combination with HMA azacitidine in treatment-naïve patients with intermediate and HR MDS (**Figure 1**)
- Endpoints:
 - Primary endpoint: OS
 - Key secondary endpoints: mOR (CR + PR + mCR)[b], overall hematological improvement, complete remission, red blood cell and platelet transfusion independence[c], PROs, OR (CR + PR), and safety

[a]28-day cycles. [b]Per investigator-assessed IWG 2006 criteria. [c]For patients who were transfusion dependent on red blood cells and/or platelet at baseline.

PATIENT DESCRIPTION AND DISPOSITION

Table 1. Demographic and Baseline Characteristics

Characteristic (N=509)	Ven + Aza (N=256)	Pbo + Aza (N=253)
Median age, years (range)	72 (31–86)	72 (25–92)
18-64 years, n (%)	53 (20.7)	64 (25.3)
≥65 years, n (%)	203 (79.3)	189 (74.7)
Male, n (%)	162 (63.3)	166 (65.6)
Race, n (%)		
White	186 (72.7)	182 (72.2)
Asian	64 (25.0)	66 (26.2)
Black or African American	5 (2.0)	4 (1.6)
Other/missing	1 (0.4)	1 (0.4)
Region, n (%)		
Europe	112 (43.8)	111 (43.9)
North America	57 (22.3)	56 (22.1)
China	26 (10.2)	27 (10.7)
Japan	25 (9.8)	23 (9.1)
Rest of world	36 (14.1)	36 (14.2)
ECOG PS		
0	97 (37.9)	101 (39.9)
1	140 (54.7)	132 (52.2)
2	19 (7.4)	20 (7.9)

Characteristic (N=509)	Ven + Aza (N=256)	Pbo + Aza (N=253)
IPSS-R prognostic risk,[a] n (%)		
Intermediate	70 (27.3)	71 (28.1)
High	96 (37.5)	99 (39.1)
Very high	90 (35.2)	83 (23.8)
Baseline eligibility for HSCT, n (%)		
Yes	50 (19.5)	47 (18.6)
No	206 (80.5)	206 (81.4)
Bone marrow blast count, n (%)		
≤5%	66 (25.8)	69 (27.3)
>5%–≤10%	93 (36.3)	91 (36.0)
>10%–≤20%	97 (37.9)	89 (35.2)
> 20%	0	4 (1.6)
Detected common mutations,[b] n (%)		
ASXL1	60 (30)	68 (33.5)
TP53	49 (24.5)	38 (18.7)
RUNX1	29 (14.5)	49 (24.1)
EZH2	5 (2.5)	13 (6.4)

Data cutoff date: April 10, 2025.
[a]From IVRS/IWRS. [b]Percentage for each mutation is based on collected (non-missing) samples. Molecular marker data are missing for 56 patients in the Ven + Aza arm and 50 patients in the Pbo + Aza arm.

- Median age was 72 years (range, 31–86) and 72 years (range, 25–92) in the Ven + Aza and Pbo + Aza arm, respectively (**Table 1**)
- Most patients had an ECOG PS of 0-1 (Ven + Aza, 237/256 [92.6%] patients and Pbo + Aza, 233/253 [92.1%] patients)
- 186/256 (72.7%) patients and 182/253 (71.9%) patients had a high to very high IPSS-R prognostic risk in the Ven + Aza and Pbo + Aza arm, respectively
- At baseline, 206/256 (80.5%) patients in the Ven + Aza arm and 206/253 (81.4%) patients in the Pbo + Aza arm were not eligible for HSCT

Figure 2. Patient Disposition

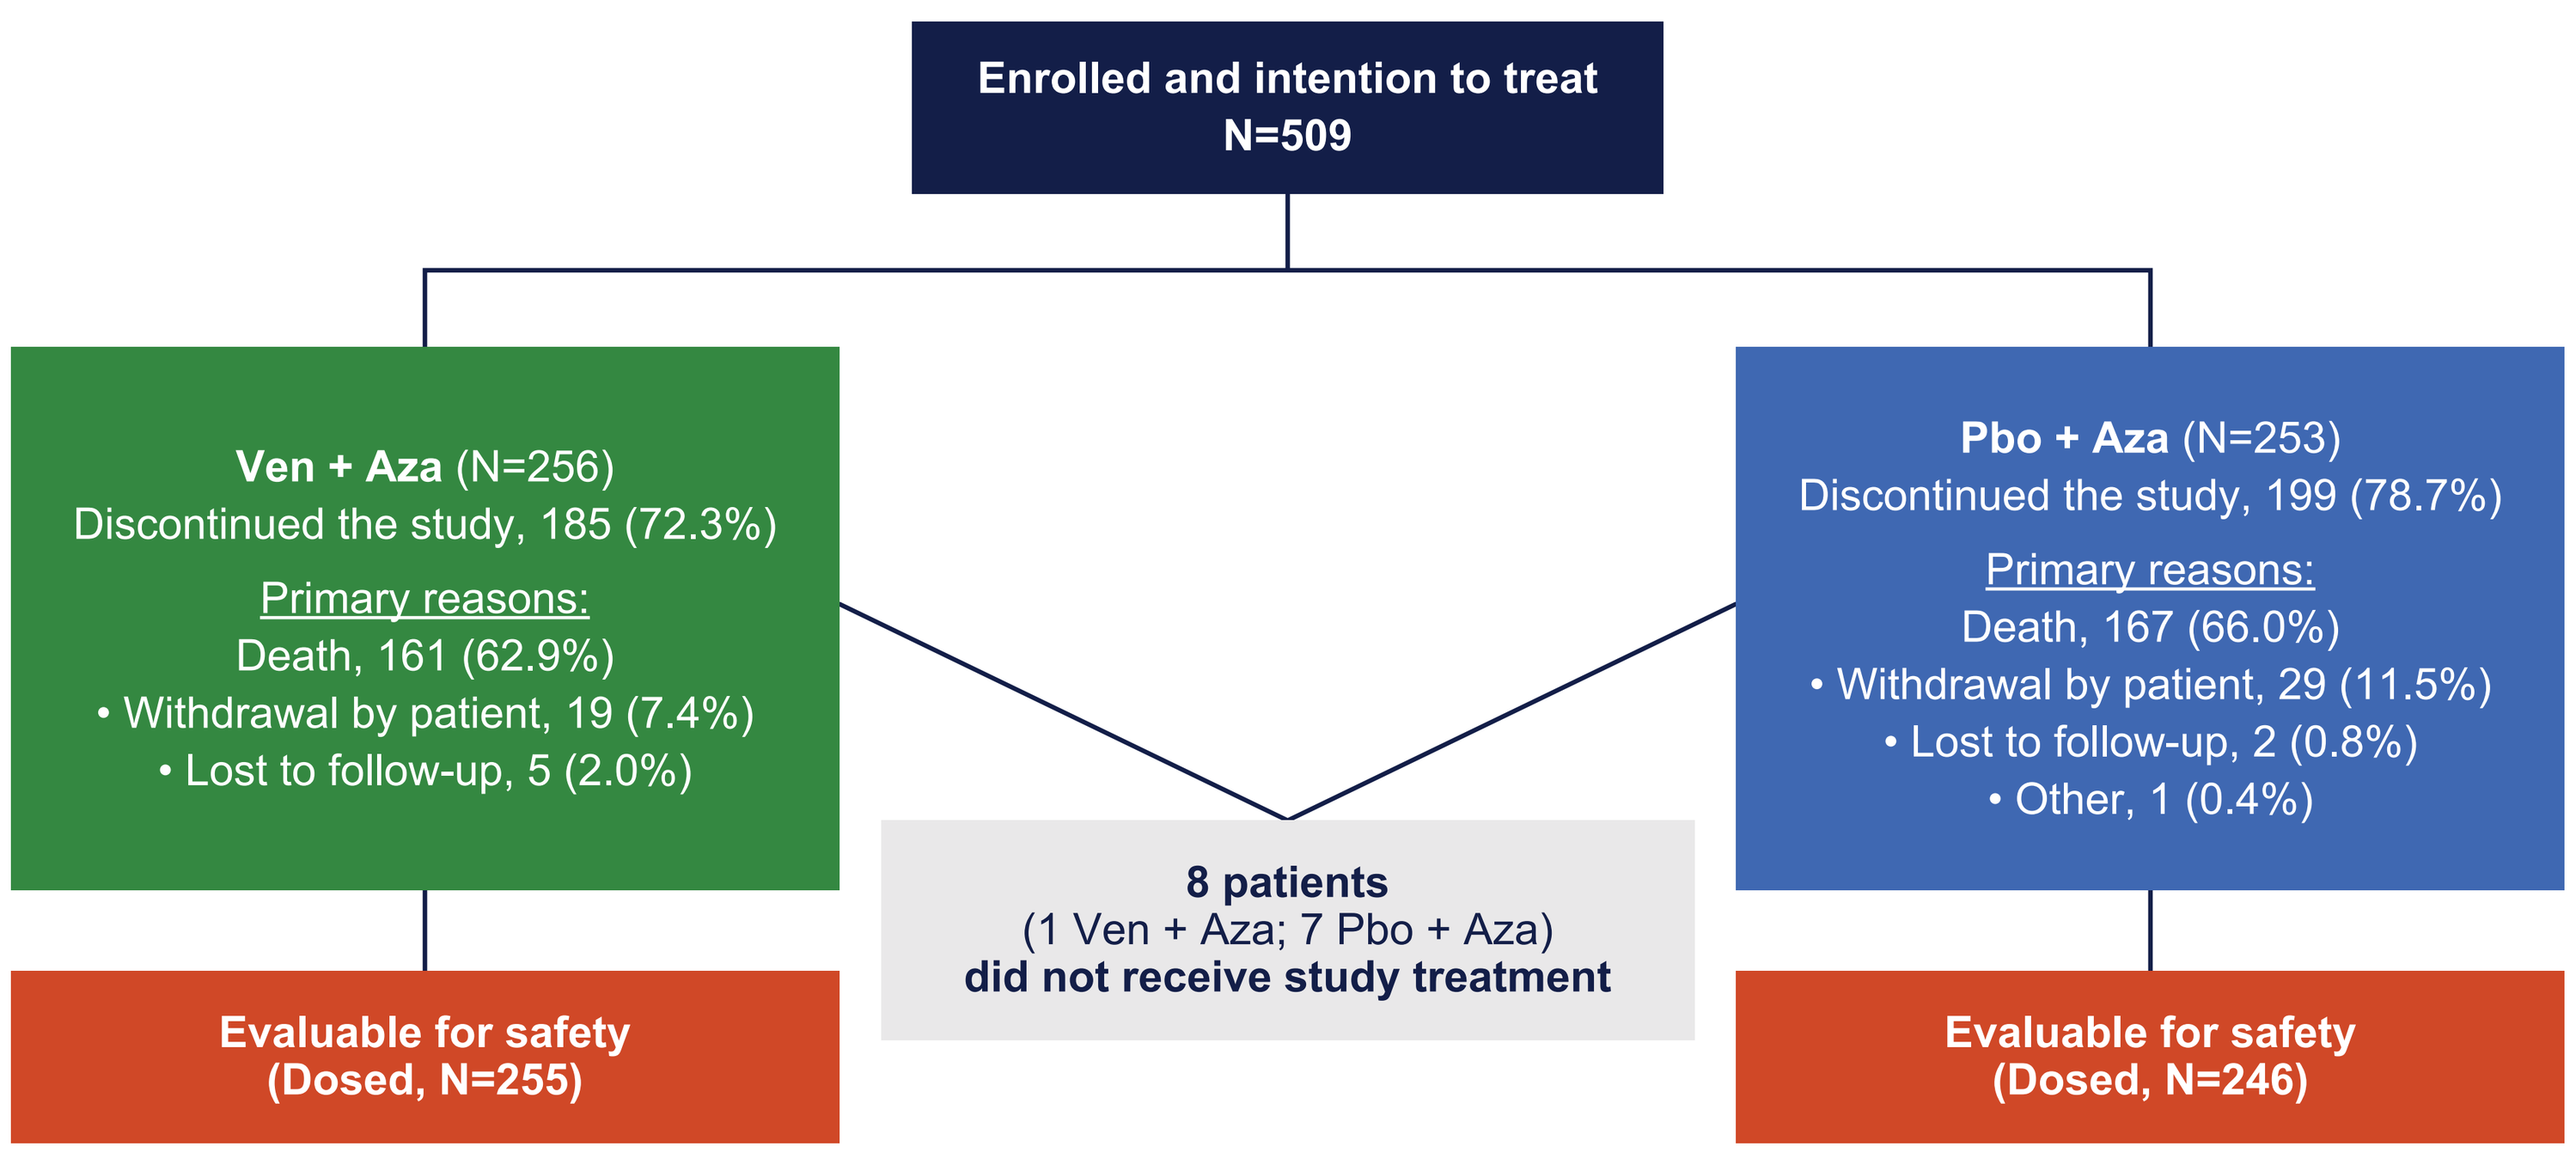
Enrolled and intention to treat N=509
Ven + Aza (N=256) Discontinued the study, 185 (72.3%)
Primary reasons:
Death, 161 (62.9%)
• Withdrawal by patient, 19 (7.4%)
• Lost to follow-up, 5 (2.0%)
Pbo + Aza (N=253) Discontinued the study, 199 (78.7%)
Primary reasons:
Death, 167 (66.0%)
• Withdrawal by patient, 29 (11.5%)
• Lost to follow-up, 2 (0.8%)
• Other, 1 (0.4%)
8 patients (1 Ven + Aza; 7 Pbo + Aza) did not receive study treatment
Evaluable for safety (Dosed, N=255)
Evaluable for safety (Dosed, N=246)

- Median follow-up was 41.2 months (range, 0.03–53.9)
- Median duration of treatment was 6.4 months (range, 0.4-54.0)[a]
- Median number of cycles was 6 (range, 1-53)
- 185/256 (72.3%) patients and 199/253 (78.7%) patients discontinued the study in the Ven + Aza and Pbo + Aza arm, respectively
 - Primary reason for discontinuation was death (Ven + Aza: 161/256 [62.9%]; Pbo + Aza: 167/253 [66.0%])
- 501 patients were evaluable for safety in the Ven + Aza (N=255) and Pbo + Aza arm (N=246), respectively

[a]Duration of treatment is calculated as min (date of the first dose in the last cycle + 27 days, date of death, cutoff date) - date of the first dose in first cycle + 1.

RESULTS

Figure 3. Overall Survival

OS[a]	Ven + Aza (N=256)	Pbo + Aza (N=253)
Median OS, months (95% CI)	22.18 (17.28–27.27)	21.68 (17.81–23.79)
HR (95% CI)	0.908 (0.733–1.126); Stratified log-rank[b], *P* value=0.38	

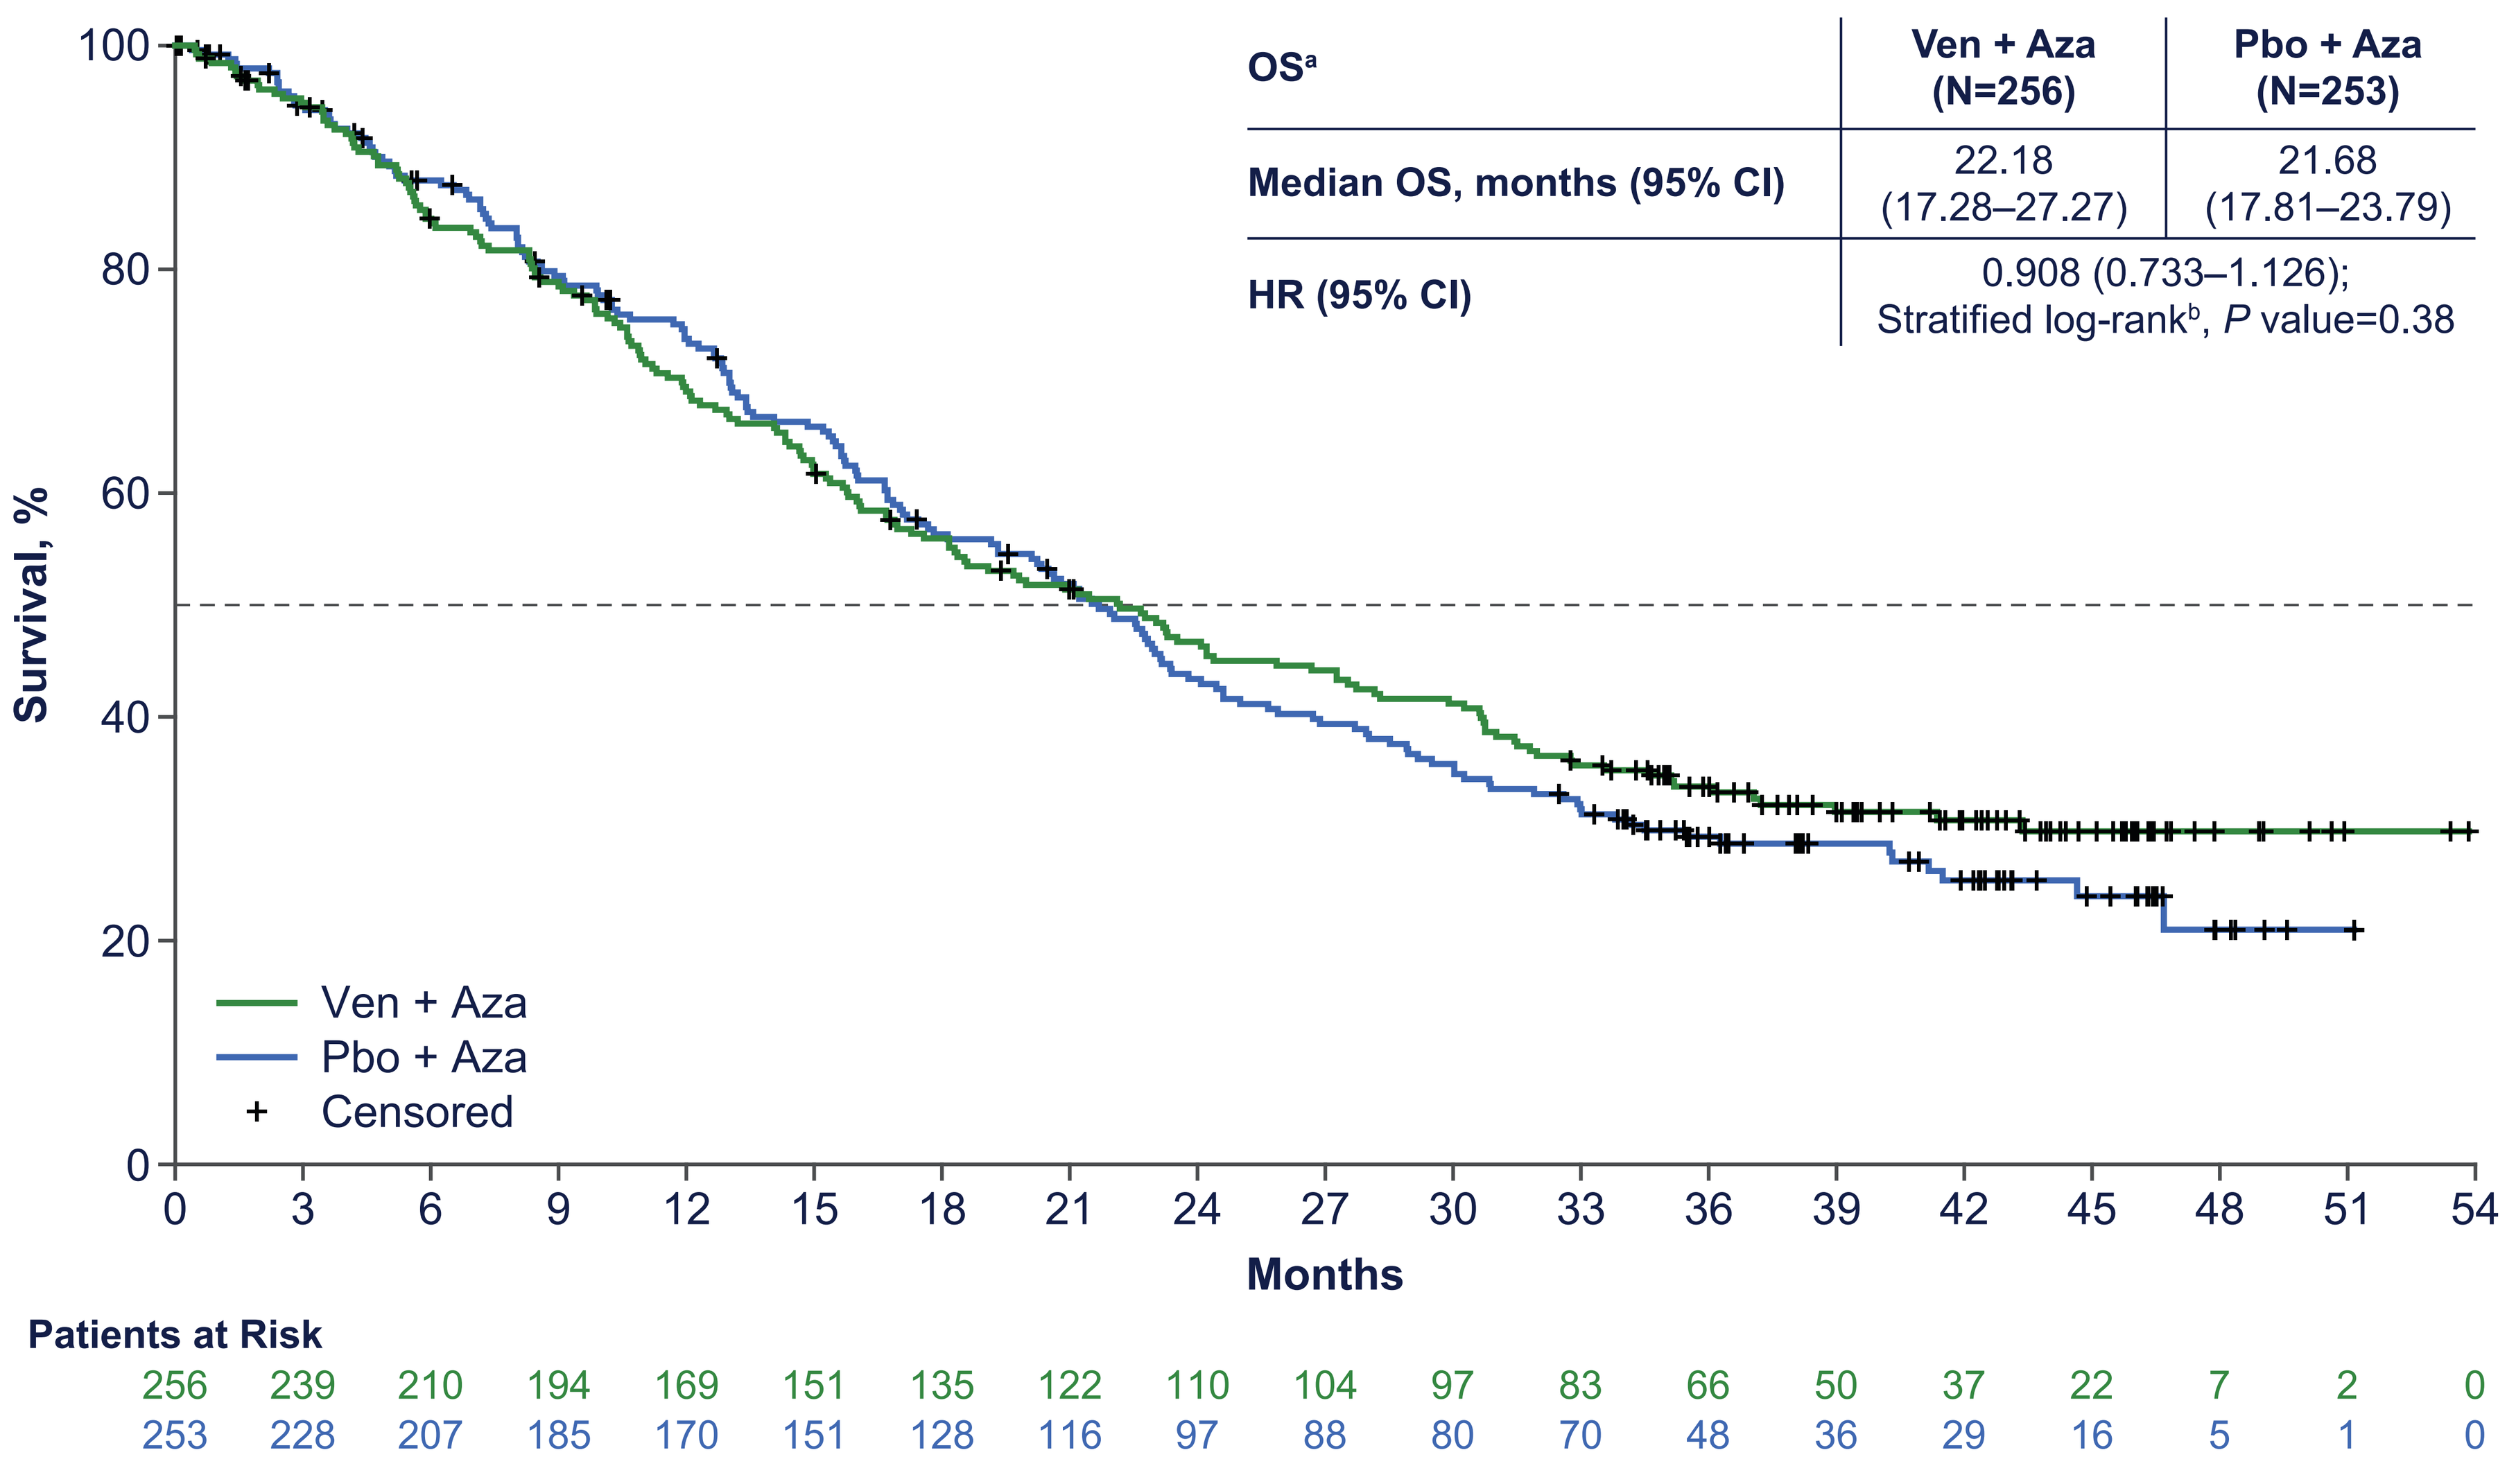
Survival, %
100
80
60
40
20
0
Ven + Aza
Pbo + Aza
+ Censored
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48 51 54
Months
Patients at Risk
256 239 210 194 169 151 135 122 110 104 97 83 66 50 37 22 7 2 0
253 228 207 185 170 151 128 116 97 88 80 70 48 36 29 16 5 1 0

Data cutoff: April 10, 2025.
[a]Overall survival was defined as the number of months from the date of the first dose of study drug to the date of death. The data were censored at the date the patient was last known to be alive on or before the cutoff date. [b]Stratified by IPSS-R (intermediate, high, very high) and HSCT eligibility (eligible, ineligible) from IVRS/IWRS.

- No difference in median overall survival was observed between Ven + Aza and Pbo + Aza in patients with intermediate and HR MDS (22.18 months [95% CI, 17.28–27.27] vs 21.68 months [95% CI, 17.81–23.79], stratified log-rank, *P* value=0.38 (**Figure 3**)

Figure 4. Modified Overall Response

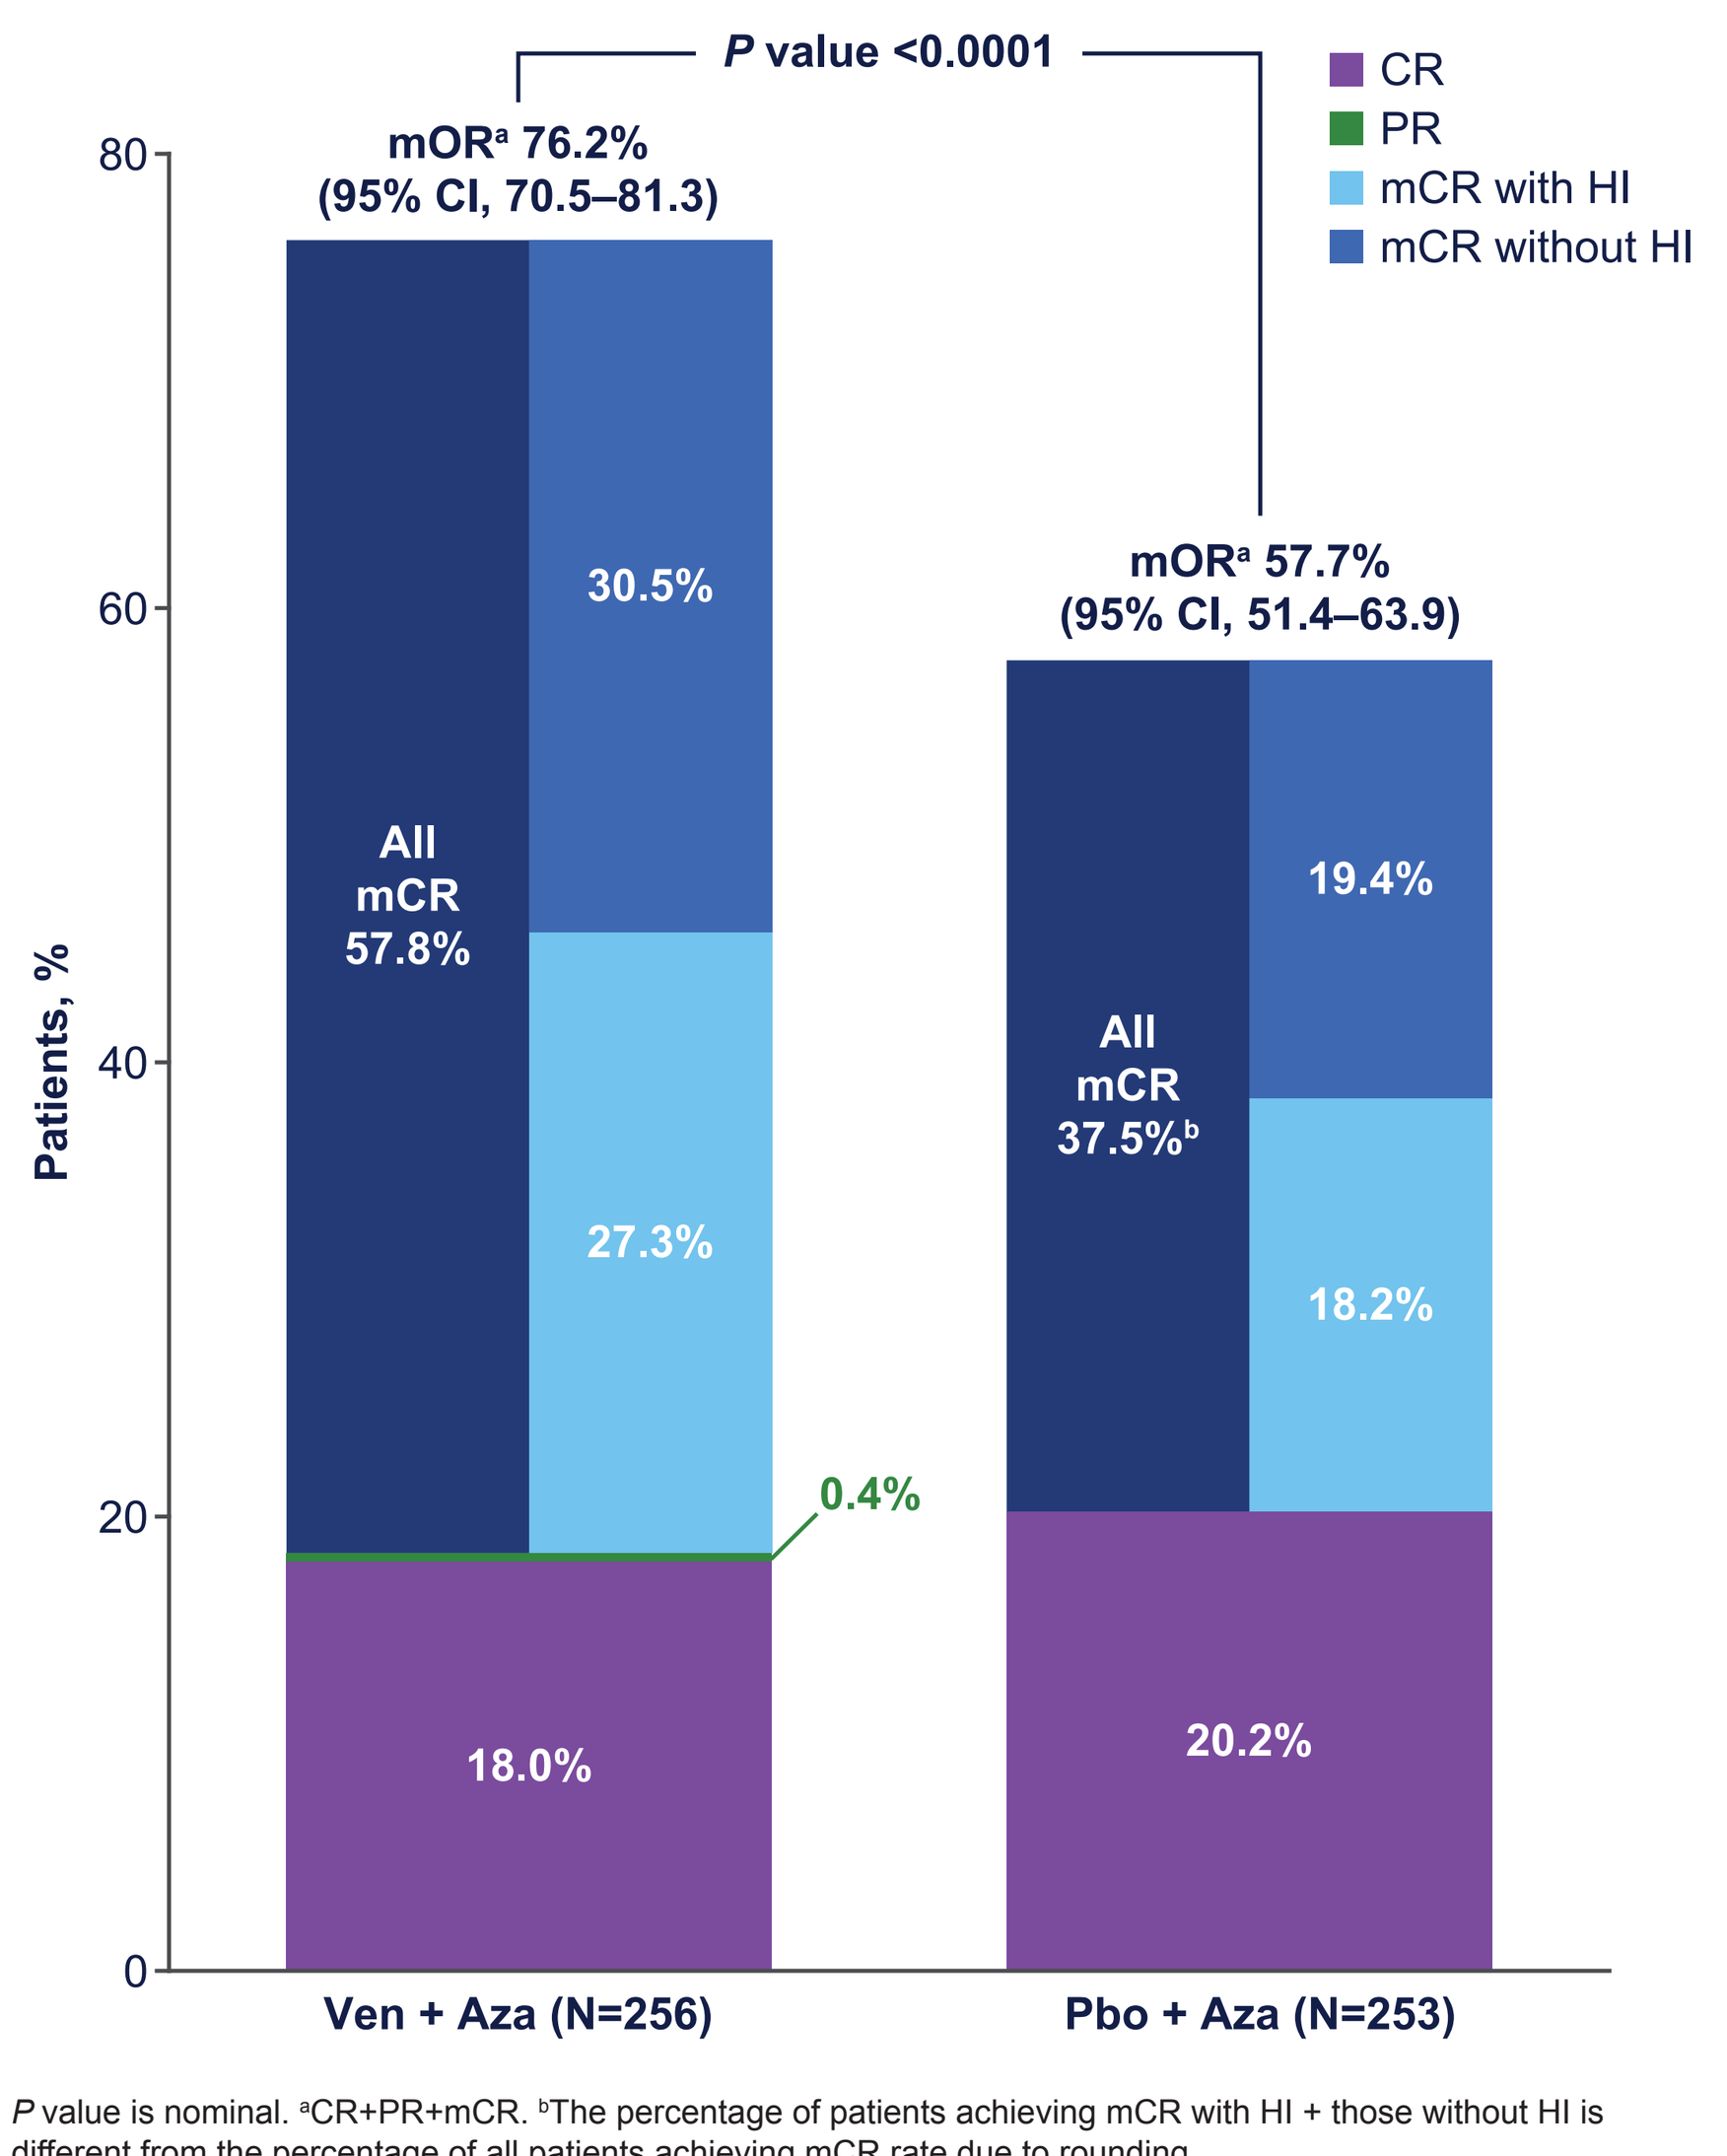
P value <0.0001
CR
PR
mCR with HI
mCR without HI
mOR[a] 76.2% (95% CI, 70.5–81.3)
mOR[a] 57.7% (95% CI, 51.4–63.9)
Patients, %
80
60
40
20
0
All mCR 57.8%
30.5%
27.3%
0.4%
18.0%
All mCR 37.5%[b]
19.4%
18.2%
20.2%
Ven + Aza (N=256)
Pbo + Aza (N=253)

P value is nominal. [a]CR+PR+mCR. [b]The percentage of patients achieving mCR with HI + those without HI is different from the percentage of all patients achieving mCR rate due to rounding.

Table 2. Overall Hematologic Improvement and Transfusion Independence Rate

	Ven + Aza (N=256)	Pbo + Aza (N=253)
Overall hematological improvement,[a] % (95% CI)	49.4 (43.0–55.8)	41.2 (35.0–47.6)
P value	0.0723	
Transfusion independence rate,[b] % (95% CI)	55.7 (47.1–64.1)	33.6 (25.7–42.1)
P value	0.0003	
Median duration of transfusion independence,[b] days (range)	183 (56-1396)	294 (59-1408)

All *P* values are nominal. [a]Based on investigator response assessment; erythroid + platelet + neutrophil responders. [b]Per the modified IWG 2006 criteria; based on patients having red blood cell or platelet transfusion within 8 weeks prior to or on first dose of study drug (red blood cell and/or platelet transfusion dependent at baseline).

- Combination of Ven + Aza resulted in a higher mOR vs Pbo + Aza (76.2% [95% CI, 70.5–81.3] vs 57.7% [95% CI, 51.4–63.9], *P*<0.0001) (**Figure 4**) and a higher transfusion independence rate (55.7% [95% CI, 47.1–64.1] vs 33.6% [95% CI, 25.7–42.1], *P* value=0.0003) (**Table 2**)

Table 3. Most Common Treatment-Emergent Adverse Events Reported in ≥20% of Patients in Either Treatment Arm

TEAEs by preferred term, n (%)	Ven + Aza (N=255)		Pbo + Aza (N=246)	
	Any grade	Grade ≥3	Any grade	Grade ≥3
Any	**253 (99.2)**	**245 (96.1)**	**244 (99.2)**	**230 (93.5)**
Neutropenia[a]	197 (77.3)	196 (76.9)	148 (60.2)	145 (58.9)
Thrombocytopenia[b]	169 (66.3)	162 (63.5)	145 (58.9)	128 (52.0)
Anemia[c]	114 (44.7)	101 (39.6)	94 (38.2)	87 (35.4)
Constipation	108 (42.4)	2 (0.8)	124 (50.4)	6 (2.4)
Diarrhea	91 (35.7)	3 (1.2)	62 (25.2)	2 (0.8)
Nausea	91 (35.7)	2 (0.8)	96 (39.0)	0
Vomiting	60 (23.5)	1 (0.4)	54 (22.0)	1 (0.4)
Febrile neutropenia	59 (23.1)	59 (23.1)	39 (15.9)	39 (15.9)
Disease progression	31 (12.2)	24 (9.4)	58 (23.6)	33 (13.4)
Pyrexia	45 (17.6)	5 (2.0)	50 (20.3)	5 (2.0)

[a]Neutropenia and neutrophil count decreased. [b]Thrombocytopenia and platelet count decreased. [c]Anemia and hemoglobin decreased.

- The most common any grade TEAEs in the Ven + Aza and Pbo + Aza arms were neutropenia (77.3% and 60.2%) and thrombocytopenia (66.3% and 58.9%) (**Table 3**)
- The most common Grade ≥3 TEAEs were neutropenia (76.9% and 58.9%) and thrombocytopenia (63.5% and 52.0%) (**Table 3**)
- No new safety signals were observed

Table 4. Summary of Safety

AEs, n (%)	Ven + Aza (N=255)	Pbo + Aza (N=246)
Serious AEs	167 (65.5)	135 (54.9)
AEs leading to		
Treatment discontinuation		
Ven/Pbo	106 (41.6)	136 (55.3)
Aza	106 (41.6)	136 (55.3)
Dose interruption		
Ven/Pbo	201 (78.8)	167 (67.9)
Aza	201 (78.8)	165 (67.1)
Dose reduction		
Ven/Pbo	32 (12.5)	17 (6.9)
Aza	124 (48.6)	67 (27.2)
AEs leading to death[a]	18 (7.1)	20 (8.1)
Disease progression	4 (1.6)	2 (0.8)
Septic shock	3 (1.2)	2 (0.8)
Sepsis	1 (0.4)	2 (0.8)
AML transformation	1 (0.4)	2 (0.8)
Cardiac arrest	1 (0.4)	3 (1.2)
Multiple organ dysfunction syndrome	1 (0.4)	2 (0.8)
Deaths	167 (65.5)	169 (68.6)
Most common cause[b]		
Disease progression	100 (39.2)	100 (40.7)
AEs	27 (10.6)	28 (11.4)
≤30 days after last dose	19 (7.5)	21 (8.5)
>30 days after last dose	147 (57.6)	147 (59.8)

[a]Death reported in ≥2 patients in either treatment arm. [b]Cause of death determined by investigator.

- AEs leading to discontinuation were lower in the Ven + Aza arm vs Pb + Aza (41.6% vs 55.3%)
- Serious AEs occurred in 65.5% of patients in the Ven + Aza arm and 54.9% of patients in the Pbo + Aza arm (**Table 4**)
- The most common cause of death was disease progression (Ven + Aza: 39.2%; Pbo + Aza: 40.7%) (**Table 4**)
- Fatal AEs were similar between treatment arms (Ven + Aza: 167; Pbo + Aza: 169)

Presented at the Society of Hematologic Oncology Annual Meeting, September 3-6, 2025, Houston, TX; DV-016300.