Poster R375

Gil Yosipovitch | yosipog@gmail.com

Adverse Events Associated With Corticosteroid Use in Chronic Spontaneous Urticaria Management

Gil Yosipovitch,[1] Dhaval Patil,[2] Jonathan Rodrigues,[2] Merin Kuruvilla,[2] Tara Raftery,[3] Irina Pivneva,[4] Jason Doran,[5] James Signorovitch,[6] Marc A. Riedl[7]

[1]Dr. Phillip Frost Department of Dermatology and Cutaneous Surgery, University of Miami Miller School of Medicine, Miami, FL, USA; [2]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [3]Novartis Ireland Ltd., Dublin, Ireland; [4]Analysis Group, Inc., Montréal, QC, Canada; [5]Analysis Group, Inc., Washington, DC, USA; [6]Analysis Group, Inc., Boston, MA, USA; [7]Division of Allergy & Immunology, Department of Medicine, University of California San Diego, La Jolla, CA, USA

KEY FINDINGS & CONCLUSIONS

- CSU-related corticosteroid use was associated with the occurrence of AEs; the most common AEs were lipid disorders, hypertension, anxiety, and obesity
- Prolonged corticosteroid use (>60 days) was observed in the management of CSU, and was typically associated with more frequent AEs and greater AE-related HCRU
- These data support restricting corticosteroids to short-term use only as a rescue treatment for managing CSU in accordance with current guidelines[1]

Scan to obtain:
- Poster

https://novartis.medicalcongressposters.com/Default.aspx?doc=9258e

Copies of this poster obtained through the Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study was funded by Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA.

Poster presented at the American College of Allergy, Asthma & Immunology Annual Scientific Meeting, November 6–10, 2025, Orlando, FL, USA.

INTRODUCTION

- CSU is characterized by the occurrence of itch, wheals, and/or angioedema lasting >6 weeks without an identifiable trigger[1]
- The recommended 1st-line treatment for CSU is second-generation H_1-AHs, whereas systemic corticosteroids are reserved only for short-term (≤10 days) use during acute flares[1]
- Frequent and/or prolonged use of systemic corticosteroids can result in serious AEs[2,3]
- Here, we describe systemic corticosteroid use and associated outcomes in patients with CSU in the US

METHODS

- This retrospective cohort study used data from the US HealthVerity health insurance claims database between January 2016 and October 2023 (**Figure 1**)
- HealthVerity data are HIPAA compliant; therefore, no IRB approval was necessary
- Patients ≥18 years of age with a confirmed diagnosis of CSU and at least 1 year of continuous enrollment prior to CSU diagnosis (allowing gaps in continuous enrollment of less than 30 days) were included
- AEs (identified using ICD-10-CM diagnosis codes) and HCRU, occurring within 1 year after the first prescription of systemic corticosteroids, were analyzed in corticosteroid users and non-users
 - Only patients using systemic corticosteroids within 15 days of CSU diagnosis were included; those with systemic corticosteroid use during the baseline period were excluded
 - For non-users, AEs and HCRU were assessed for 1 year after a reference date derived from corticosteroid users
- Corticosteroid users were categorized by total days of systemic corticosteroid supply as a proxy for duration of use: ≤31 days (≤31D), >31 and ≤60 days (>31–≤60D), and >60 days (>60D)
- *P* values were generated from univariable binomial logistic regression models and refer to statistical comparisons of the frequency of AEs between different corticosteroid user groups vs non-users

Figure 1. Study Design

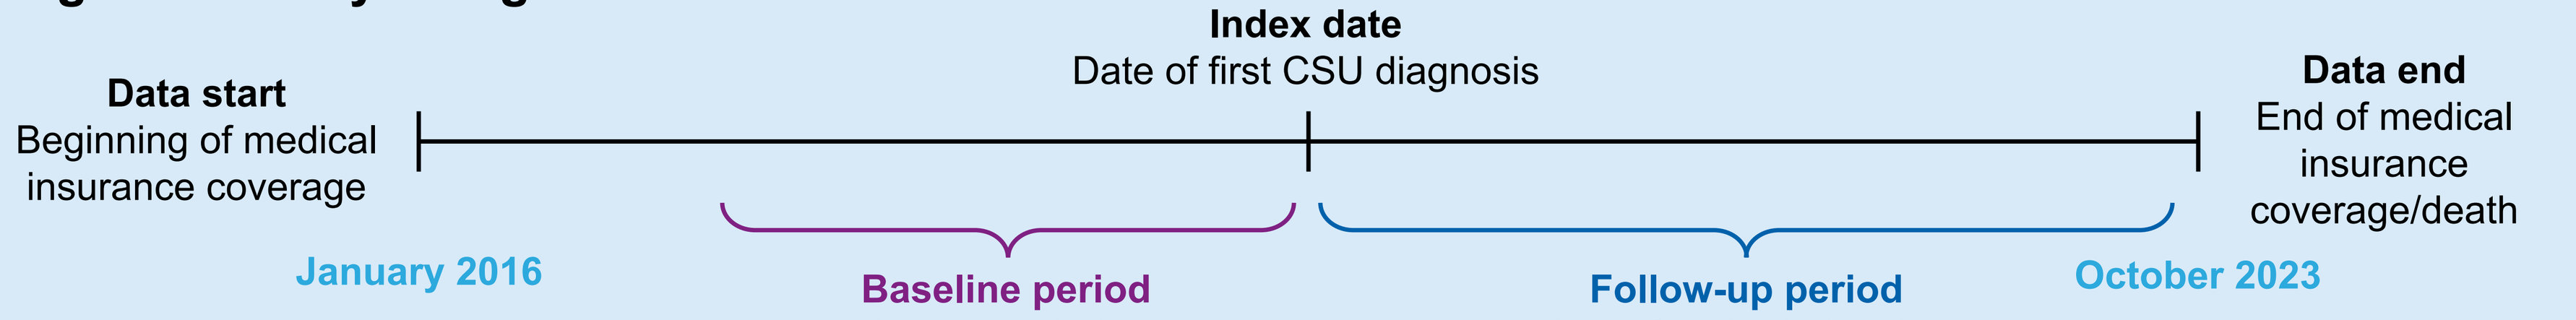
Index date
Date of first CSU diagnosis
Data start
Beginning of medical insurance coverage
Data end
End of medical insurance coverage/death
January 2016
Baseline period
1 year pre-index
Follow-up period
Time after the index date
October 2023

CSU, chronic spontaneous urticaria.

RESULTS

Patient Selection and Baseline Characteristics

- Of the 224,958 patients in this study, a total of 174,555 (77.6%) were prescribed systemic corticosteroids: 96,816 (43.0%) during the baseline period and 153,679 (68.3%) following CSU diagnosis
 - The mean (SD) number of episodes of systemic corticosteroid use was 2.1 (1.6), and the duration of use was 82.6 (143.2) days
- Overall, 87,284 met the inclusion criteria for this analysis, of whom 42,478 were corticosteroid users and 44,806 were non-corticosteroid users (**Figure 2**)
- The mean age at the index date was 43.3 and 44.3 years for corticosteroid users and non-users, respectively, and most patients were female (**Table 1**)

Figure 2. Patient Selection

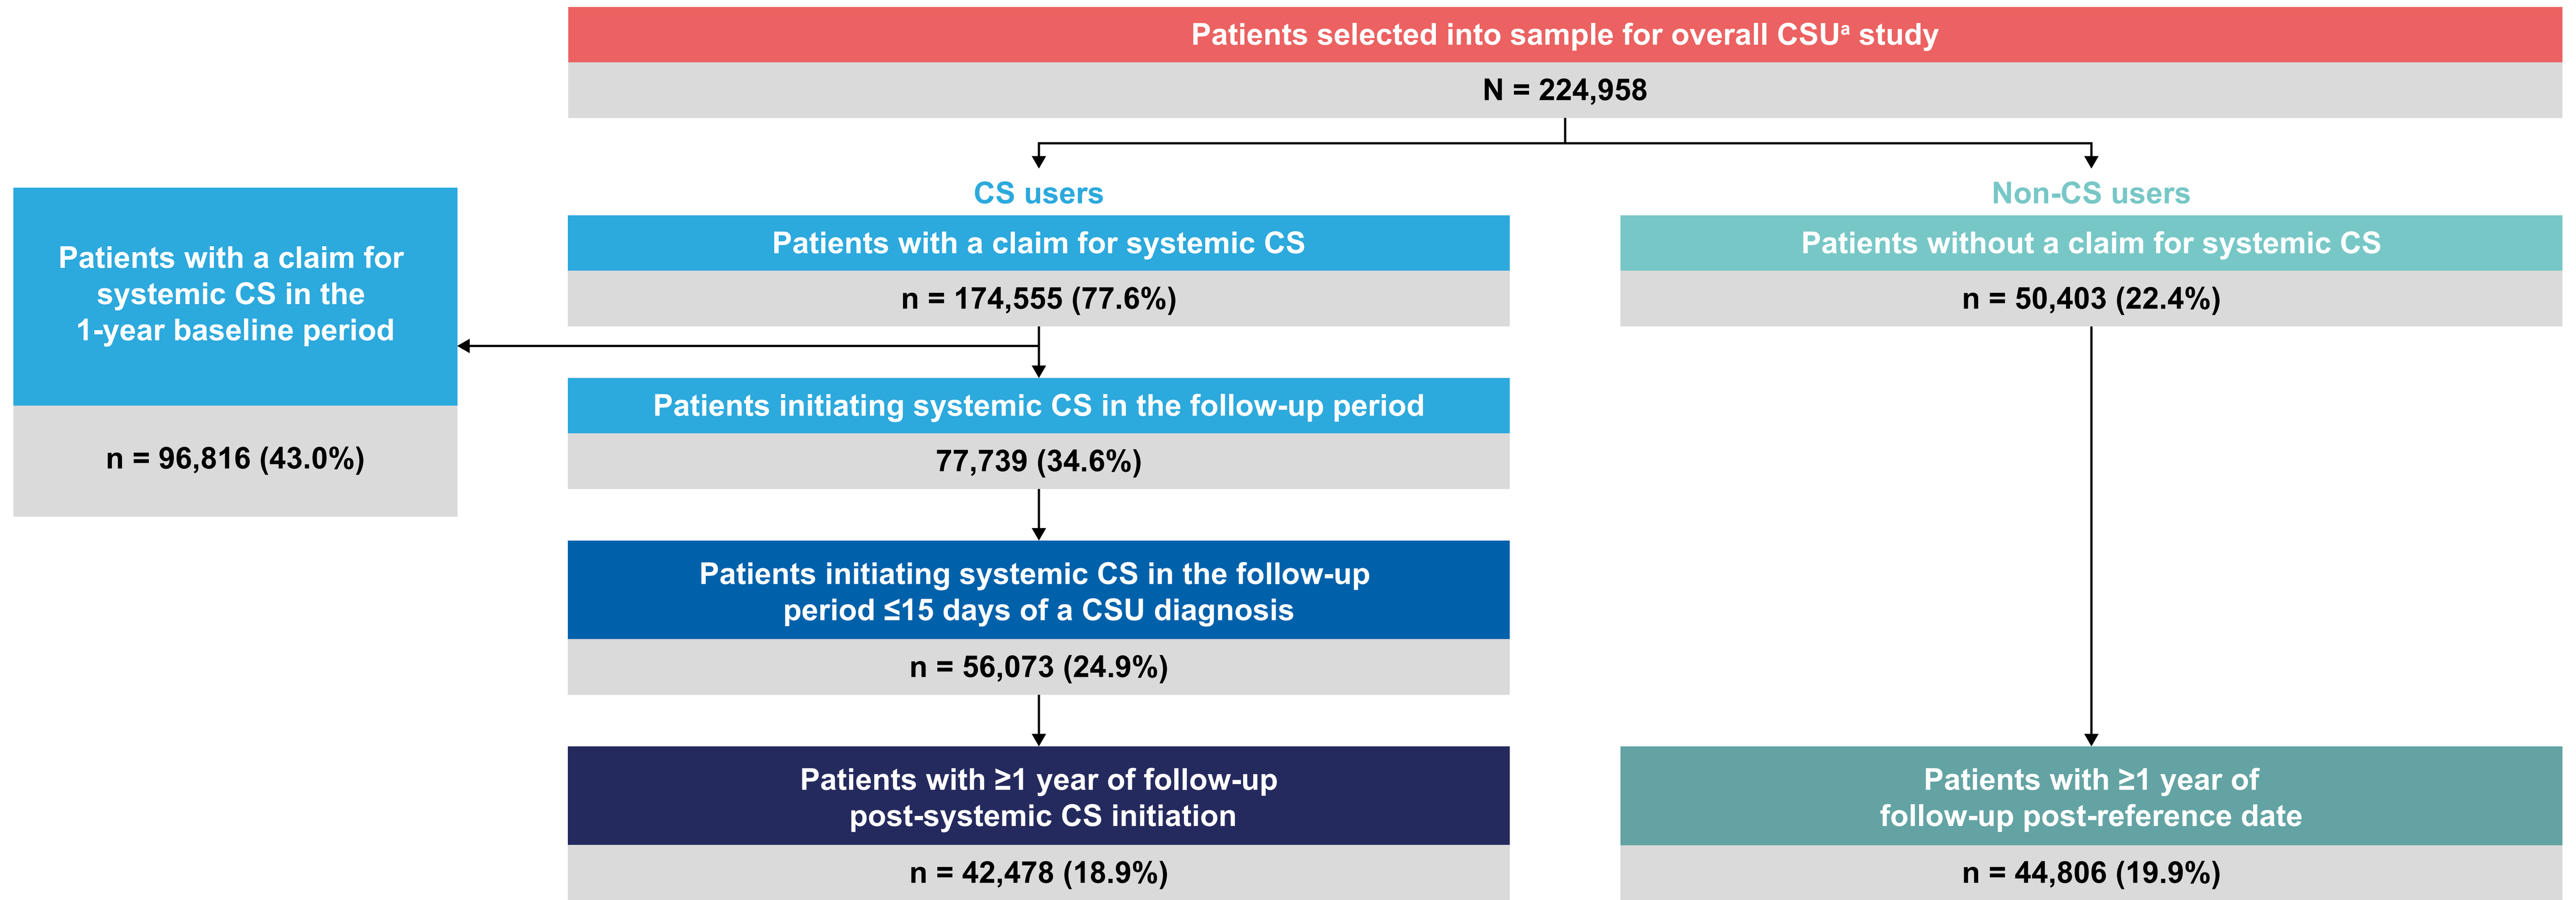
Patients selected into sample for overall CSU[a] study
N = 224,958
CS users
Non-CS users
Patients with a claim for systemic CS in the 1-year baseline period
n = 96,816 (43.0%)
Patients with a claim for systemic CS
n = 174,555 (77.6%)
Patients without a claim for systemic CS
n = 50,403 (22.4%)
Patients initiating systemic CS in the follow-up period
77,739 (34.6%)
Patients initiating systemic CS in the follow-up period ≤15 days of a CSU diagnosis
n = 56,073 (24.9%)
Patients with ≥1 year of follow-up post-systemic CS initiation
n = 42,478 (18.9%)
Patients with ≥1 year of follow-up post-reference date
n = 44,806 (19.9%)

[a]A confirmed diagnosis for CSU was defined as ≥2 diagnoses of either idiopathic, other, or unspecified urticaria or ≥1 diagnosis of idiopathic, other, or unspecified urticaria and ≥1 diagnosis of angioedema separated by ≥6 weeks but ≤1 year apart, in either order.
CS, corticosteroid(s); CSU, chronic spontaneous urticaria.

Table 1. Patient Characteristics

Patient characteristics	Non-users n = 44,806	All CS users n = 42,478	All CS users (n = 42,478): ≤31D (n = 39,510)	>31–≤60D (n = 2104)	>60D (n = 864)
Age at index date,[a] years, mean (SD)	44.3 (16.6)	43.3 (15.4)	43.1 (15.4)	45.5 (14.3)	48.6 (14.3)
Female sex, n (%)	32,592 (72.7)	31,656 (74.5)	29,584 (74.9)	1466 (69.7)	606 (70.1)
Payer type,[b] n (%)					
Commercial	29,420 (65.7)	27,751 (65.3)	25,605 (64.8)	1514 (72.0)	632 (73.1)
Medicaid	12,307 (27.5)	12,126 (28.5)	11,504 (29.1)	465 (22.1)	157 (18.2)
Medicare Advantage	2717 (6.1)	2255 (5.3)	2081 (5.3)	106 (5.0)	68 (7.9)
Unknown	362 (0.8)	346 (0.8)	320 (0.8)	19 (0.9)	7 (0.8)
CCI, mean (SD)	0.5 (1.1)	0.4 (1.0)	0.4 (1.0)	0.4 (1.0)	0.5 (1.1)
Duration of follow-up period, years, mean (SD)	2.7 (1.3)	3.0 (1.5)	3.0 (1.5)	3.1 (1.5)	3.2 (1.5)

[a]The index date was specified as the earliest of the two diagnosis dates used to confirm CSU. [b]When multiple payer types were reported, commercial insurance was prioritized, followed by Medicaid, and then Medicare Advantage.
Patients were categorized according to total days of systemic CS supply in the 1 year after the first systemic CS prescription (or reference date) during the follow-up period.
CCI, Charlson Comorbidity Index; CS, corticosteroid(s); CSU, chronic spontaneous urticaria; D, days; SD, standard deviation.

Limitations

- Dosage and potency of corticosteroids were not considered in this analysis
- Changes in AE-related HCRU were assessed as a whole; therefore, the weighted attribution of specific AEs could not be ascertained

Adverse Events

- Among the 87,284 patients, the most common AEs included lipid disorders, hypertension, and anxiety (**Figure 3**)
- AEs typically occurred in a higher proportion of corticosteroid users relative to non-users
- Generally, the proportion of patients with AEs increased with increasing time on corticosteroids

Figure 3. Most Common AEs Within 1 Year After the First Prescription of Systemic CS Following CSU Diagnosis

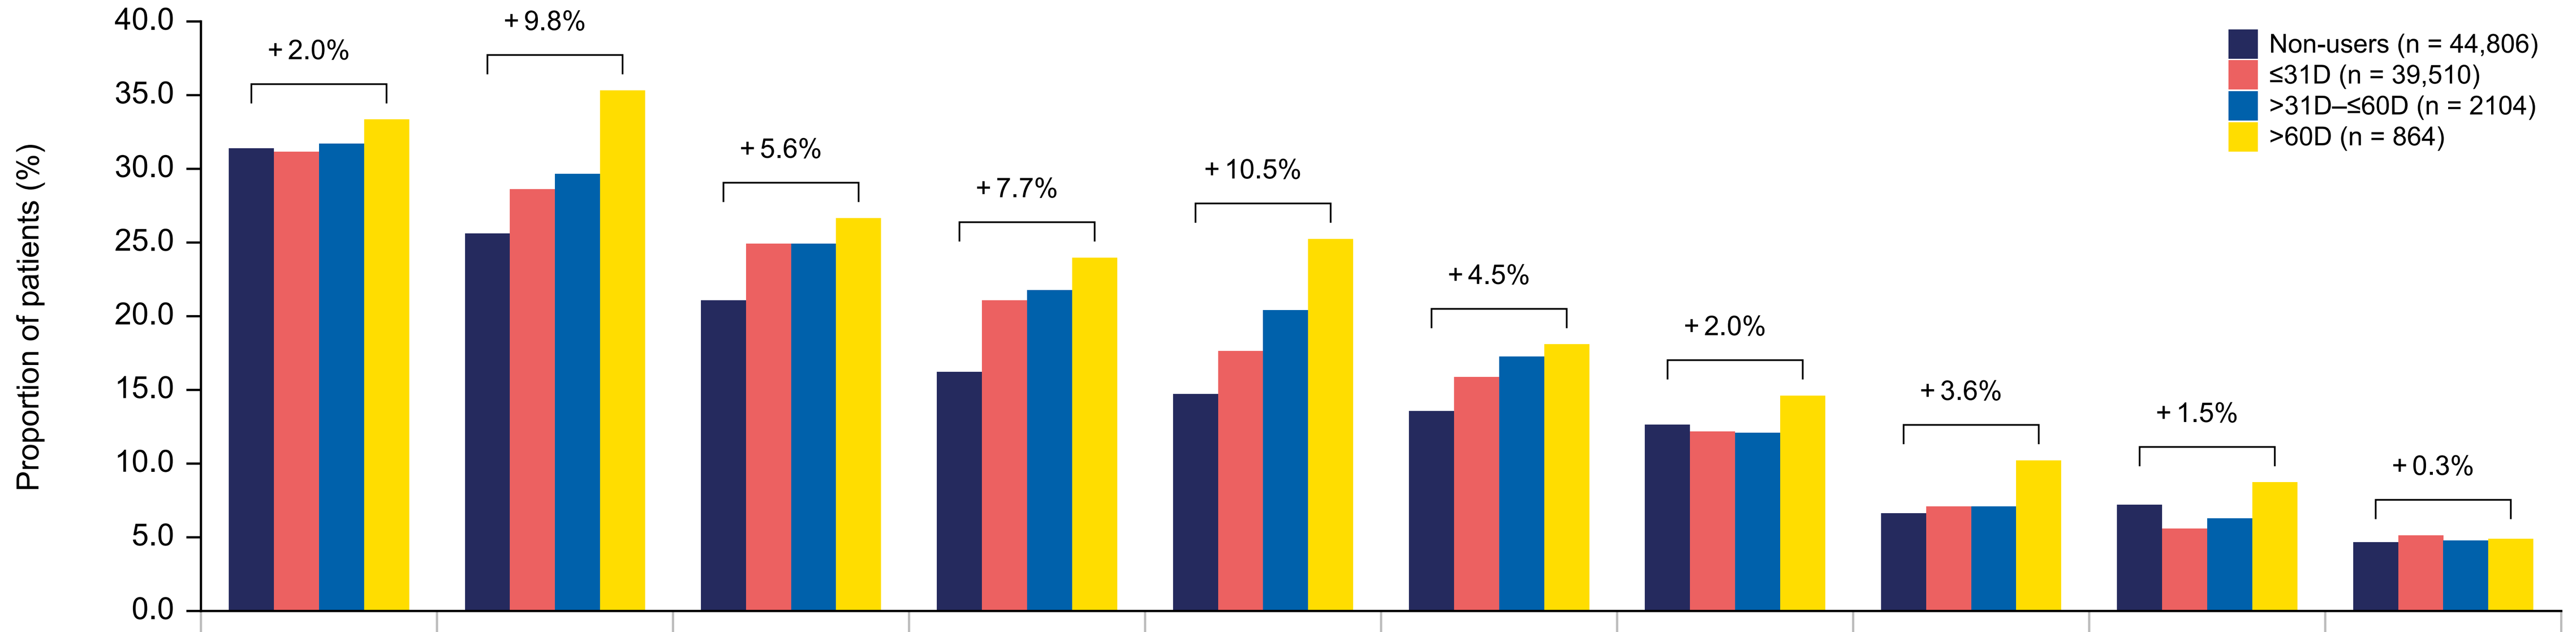
Proportion of patients (%)
40.0
35.0
30.0
25.0
20.0
15.0
10.0
5.0
0.0
+2.0%
+9.8%
+5.6%
+7.7%
+10.5%
+4.5%
+2.0%
+3.6%
+1.5%
+0.3%
Non-users (n = 44,806)
≤31D (n = 39,510)
>31D–≤60D (n = 2104)
>60D (n = 864)

	Lipid disorders	Hypertension	Anxiety	Obesity	Fatigue	Depression	Diabetes	Insomnia	Cataracts	Gastritis
Non-users (n = 44,806)	31.4%	25.6%	21.1%	16.3%	14.8%	13.6%	12.7%	6.7%	7.3%	4.7%
≤31D (n = 39,150)	31.2% (*P*=0.467)	28.7% (*P*<0.001)	25.0% (*P*<0.001)	21.2% (*P*<0.001)	17.7% (*P*<0.001)	15.9% (*P*<0.001)	12.2% (*P*=0.064)	7.1% (*P*=0.050)	5.7% (*P*<0.001)	5.2% (*P*<0.001)
>31D–≤60D (n = 2104)	31.8% (*P*=0.701)	29.7% (*P*<0.001)	25.0% (*P*<0.001)	21.8% (*P*<0.001)	20.5% (*P*<0.001)	17.3% (*P*<0.001)	12.1% (*P*=0.467)	7.1% (*P*=0.507)	6.4% (*P*=0.121)	4.8% (*P*=0.755)
>60D (n = 864)	33.4% (*P*=0.199)	35.4% (*P*<0.001)	26.7% (*P*<0.001)	24.0% (*P*<0.001)	25.3% (*P*<0.001)	18.1% (*P*<0.001)	14.7% (*P*=0.076	10.3% (*P*<0.001)	8.8% (*P*=0.087)	5.0% (*P*=0.706)
All CS users (n = 42,478)	31.2%	28.9%	25.0%	21.3%	18.0%	16.0%	12.3%	7.1%	5.8%	5.2%

Patients were categorized according to total days of systemic CS supply in the 1 year after the first systemic CS prescription (or reference date) during the follow-up period. Data for the "All CS users" group is not displayed on the bar chart. Numerical differences between the "Non-users" and ">60D" groups are shown on the chart for each AE.
AE, adverse event; CS, corticosteroid(s); CSU, chronic spontaneous urticaria; D, days.

Adverse Event-Related Health Care Resource Utilization

- AE-related outpatient visits, inpatient admissions, and ED visits increased with longer corticosteroid use (**Figure 4**)

Figure 4. AE-Related HCRU During Follow-Up PPPY

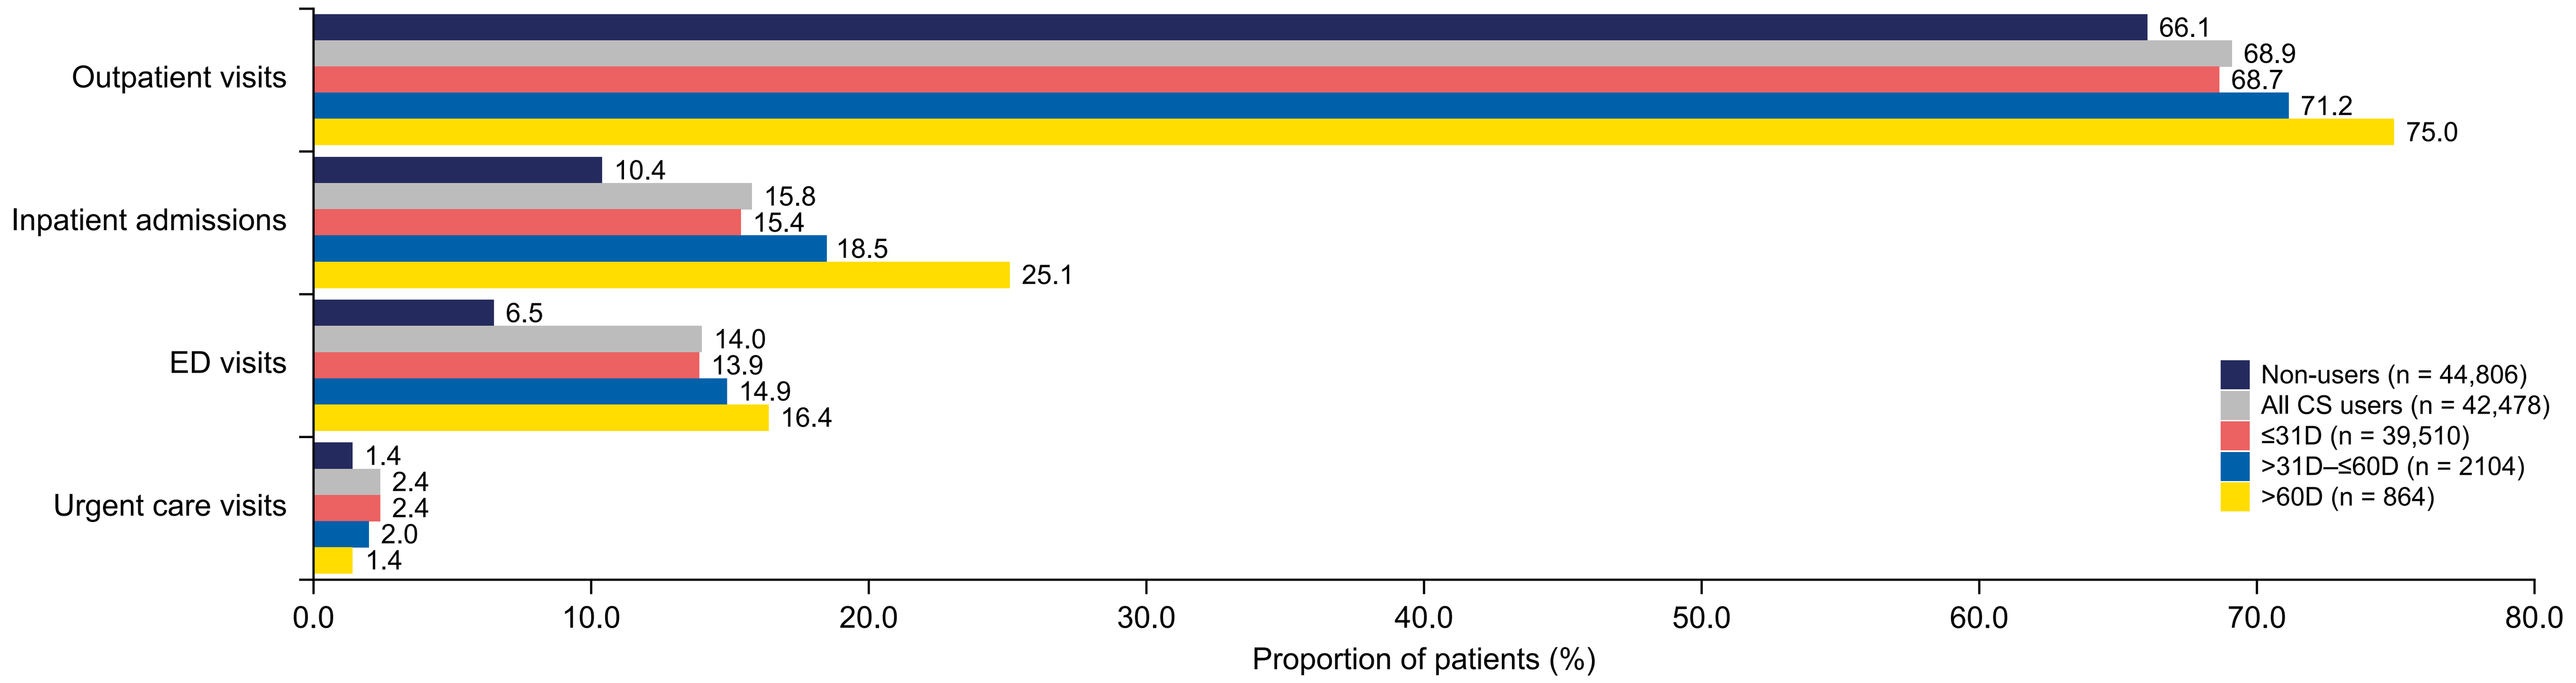
Outpatient visits
66.1
68.9
68.7
71.2
75.0
Inpatient admissions
10.4
15.8
15.4
18.5
25.1
ED visits
6.5
14.0
13.9
14.9
16.4
Urgent care visits
1.4
2.4
2.4
2.0
1.4
0.0
10.0
20.0
30.0
40.0
50.0
60.0
70.0
80.0
Proportion of patients (%)
Non-users (n = 44,806)
All CS users (n = 42,478)
≤31D (n = 39,510)
>31D–≤60D (n = 2104)
>60D (n = 864)

Patients were categorized according to total days of systemic CS supply in the 1 year after the first systemic CS prescription (or reference date) during the follow-up period.
Patients with other visits are not displayed on the graph. Other visits included causes where a visit could not be identified as any of the prespecified categories of visit.
AE, adverse event; CS, corticosteroid(s); D, days; ED, emergency department; HCRU, health care resource utilization; PPPY, per patient per year.

References

1. Zuberbier T, et al. *Allergy.* 2022;77(3):734–766.
2. Ledford D, et al. *Allergy Asthma Proc.* 2016;37(6):458–465.
3. Waljee AK, et al. *BMJ.* 2017;357:j1415.

Abbreviations

AE, adverse event; CCI, Charlson Comorbidity Index; CS, corticosteroid(s); CSU, chronic spontaneous urticaria; D, days; ED, emergency department; H_1-AH, H_1-antihistamine; HCRU, health care resource utilization; HIPAA, Health Insurance Portability and Accountability Act; ICD-10-CM, International Classification of Diseases, Tenth Revision, Clinical Modification; IRB, institutional review board; PPPY, per patient per year; SD, standard deviation; US, United States.

Acknowledgments

Writing support was provided by Hayley Loy, PhD (BOLDSCIENCE Ltd., UK), and was funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP) guidelines. The authors had full control of the content and made the final decision on all aspects of this publication.

Disclosures

GY has received honoraria as a consultant and/or advisory board member for AbbVie, Amgen, Arcutis Biotherapeutics, Celldex, CSL Vifor, Eli Lilly, Escient Pharmaceuticals, Galderma, GSK, Kamari Pharma, Kiniksa Pharmaceuticals, LEO Pharma, Maruho, Novartis, Pfizer, Pierre Fabre, Regeneron Pharmaceuticals, Inc., Sanofi, and Trevi Therapeutics; has received research funding from Celldex, Eli Lilly, Escient Health, Kiniksa Pharmaceuticals, Novartis, Pfizer, Regeneron Pharmaceuticals, Inc., and Sanofi. **DP**, **JR**, **MK**, and **TR** are full-time employees of Novartis and may own stock or stock options. **IP**, **JD**, and **JS** are full-time employees of Analysis Group, Inc., a consulting company that has provided paid consulting services to Novartis Pharma AG. **MAR** has received research and/or consulting support from Astria Therapeutics, BioCryst, BioMarin, Celldex, CSL Behring, Cycle Pharmaceuticals, Grifols, Intellia Therapeutics, Ionis Pharmaceuticals, KalVista Pharmaceuticals, Novartis, Pfizer, Pharming, Pharvaris, Sanofi-Regeneron, and Takeda Pharmaceuticals.